Multiple Proteins Correlate With Tryptase Levels in Patients With Indolent Systemic Mastocytosis (ISM): Preliminary Results of Plasma Proteomic Analysis in PIONEER

Poster Number 518

Vito Sabato,[1] Jens Panse,[2,3] Tracy I. George,[4] Hussein A. Abbas,[5] Min Chen,[6] Guang Yang,[6] Rachel L. Erlich,[6] Benjamin Lampson,[6] Jason Gotlib[7]

[1]University of Antwerp and Antwerp University Hospital, Antwerp, Belgium; [2]Department of Oncology, Hematology, Hemostaseology, and Stem Cell Transplantation, University Hospital Aachen, Medical Faculty, RWTH Aachen University, Aachen, Germany; [3]Center for Integrated Oncology (CIO), Aachen, Bonn, Cologne, Düsseldorf (ABCD), Aachen, Germany; [4]ARUP Laboratories, Huntsman Cancer Institute, Department of Pathology, University of Utah School of Medicine, Salt Lake City, UT; [5]Department of Leukemia, Division of Cancer Medicine, MD Anderson Cancer Center, Houston, TX; [6]Blueprint Medicines Corporation, Cambridge, MA; [7]Stanford Cancer Institute/Stanford University School of Medicine, Stanford, CA

Introduction

- The prevalence of systemic mastocytosis (SM) has been estimated at up to 1 in 5,000 people[1]
- Indolent systemic mastocytosis (ISM), the most common subtype of SM, is a clonal mast cell (MC) disease driven by the *KIT* D816V mutation in ~95% of patients[2–5]
- Patients with ISM often experience life-long debilitating symptoms that considerably impact quality of life[6–9]
 - These symptoms are likely due to a combination of direct MC organ infiltration, MC activation and mediator release, and inflammatory immune system alterations promoted by aberrant MCs[9]
- Tryptase plays an important role in inflammation and serves as a marker of MC burden and MC activation[10]
- Serum tryptase >20 ng/mL is a minor diagnostic criterion for SM per WHO classification.[11] However, ~30% of patients with ISM do not meet this criterion and baseline serum tryptase does not always correlate well with disease symptoms[12]
- Avapritinib is a potent, oral inhibitor that selectively targets the KIT D816V mutation,[13] with approximately 30-fold selectivity for the mutant KIT enzyme over the wild-type KIT enzyme[14]
- Avapritinib was approved in the USA and Europe for the treatment of moderate-to-severe ISM, based on the findings from the randomized, double-blind, placebo-controlled PIONEER study (NCT03731260)[15,16]
- To identify measurable disease-related biomarkers, we conducted high-throughput plasma proteome profiling on samples from patients with ISM enrolled in PIONEER to assess correlations between inflammatory plasma proteins and tryptase

Methods

- The Olink® Explore 384 Inflammation panel (Uppsala, Sweden) measured 363 well-established inflammatory plasma proteins in plasma samples from patients in PIONEER and healthy donors (**Figure 1**)

Figure 1. Study design

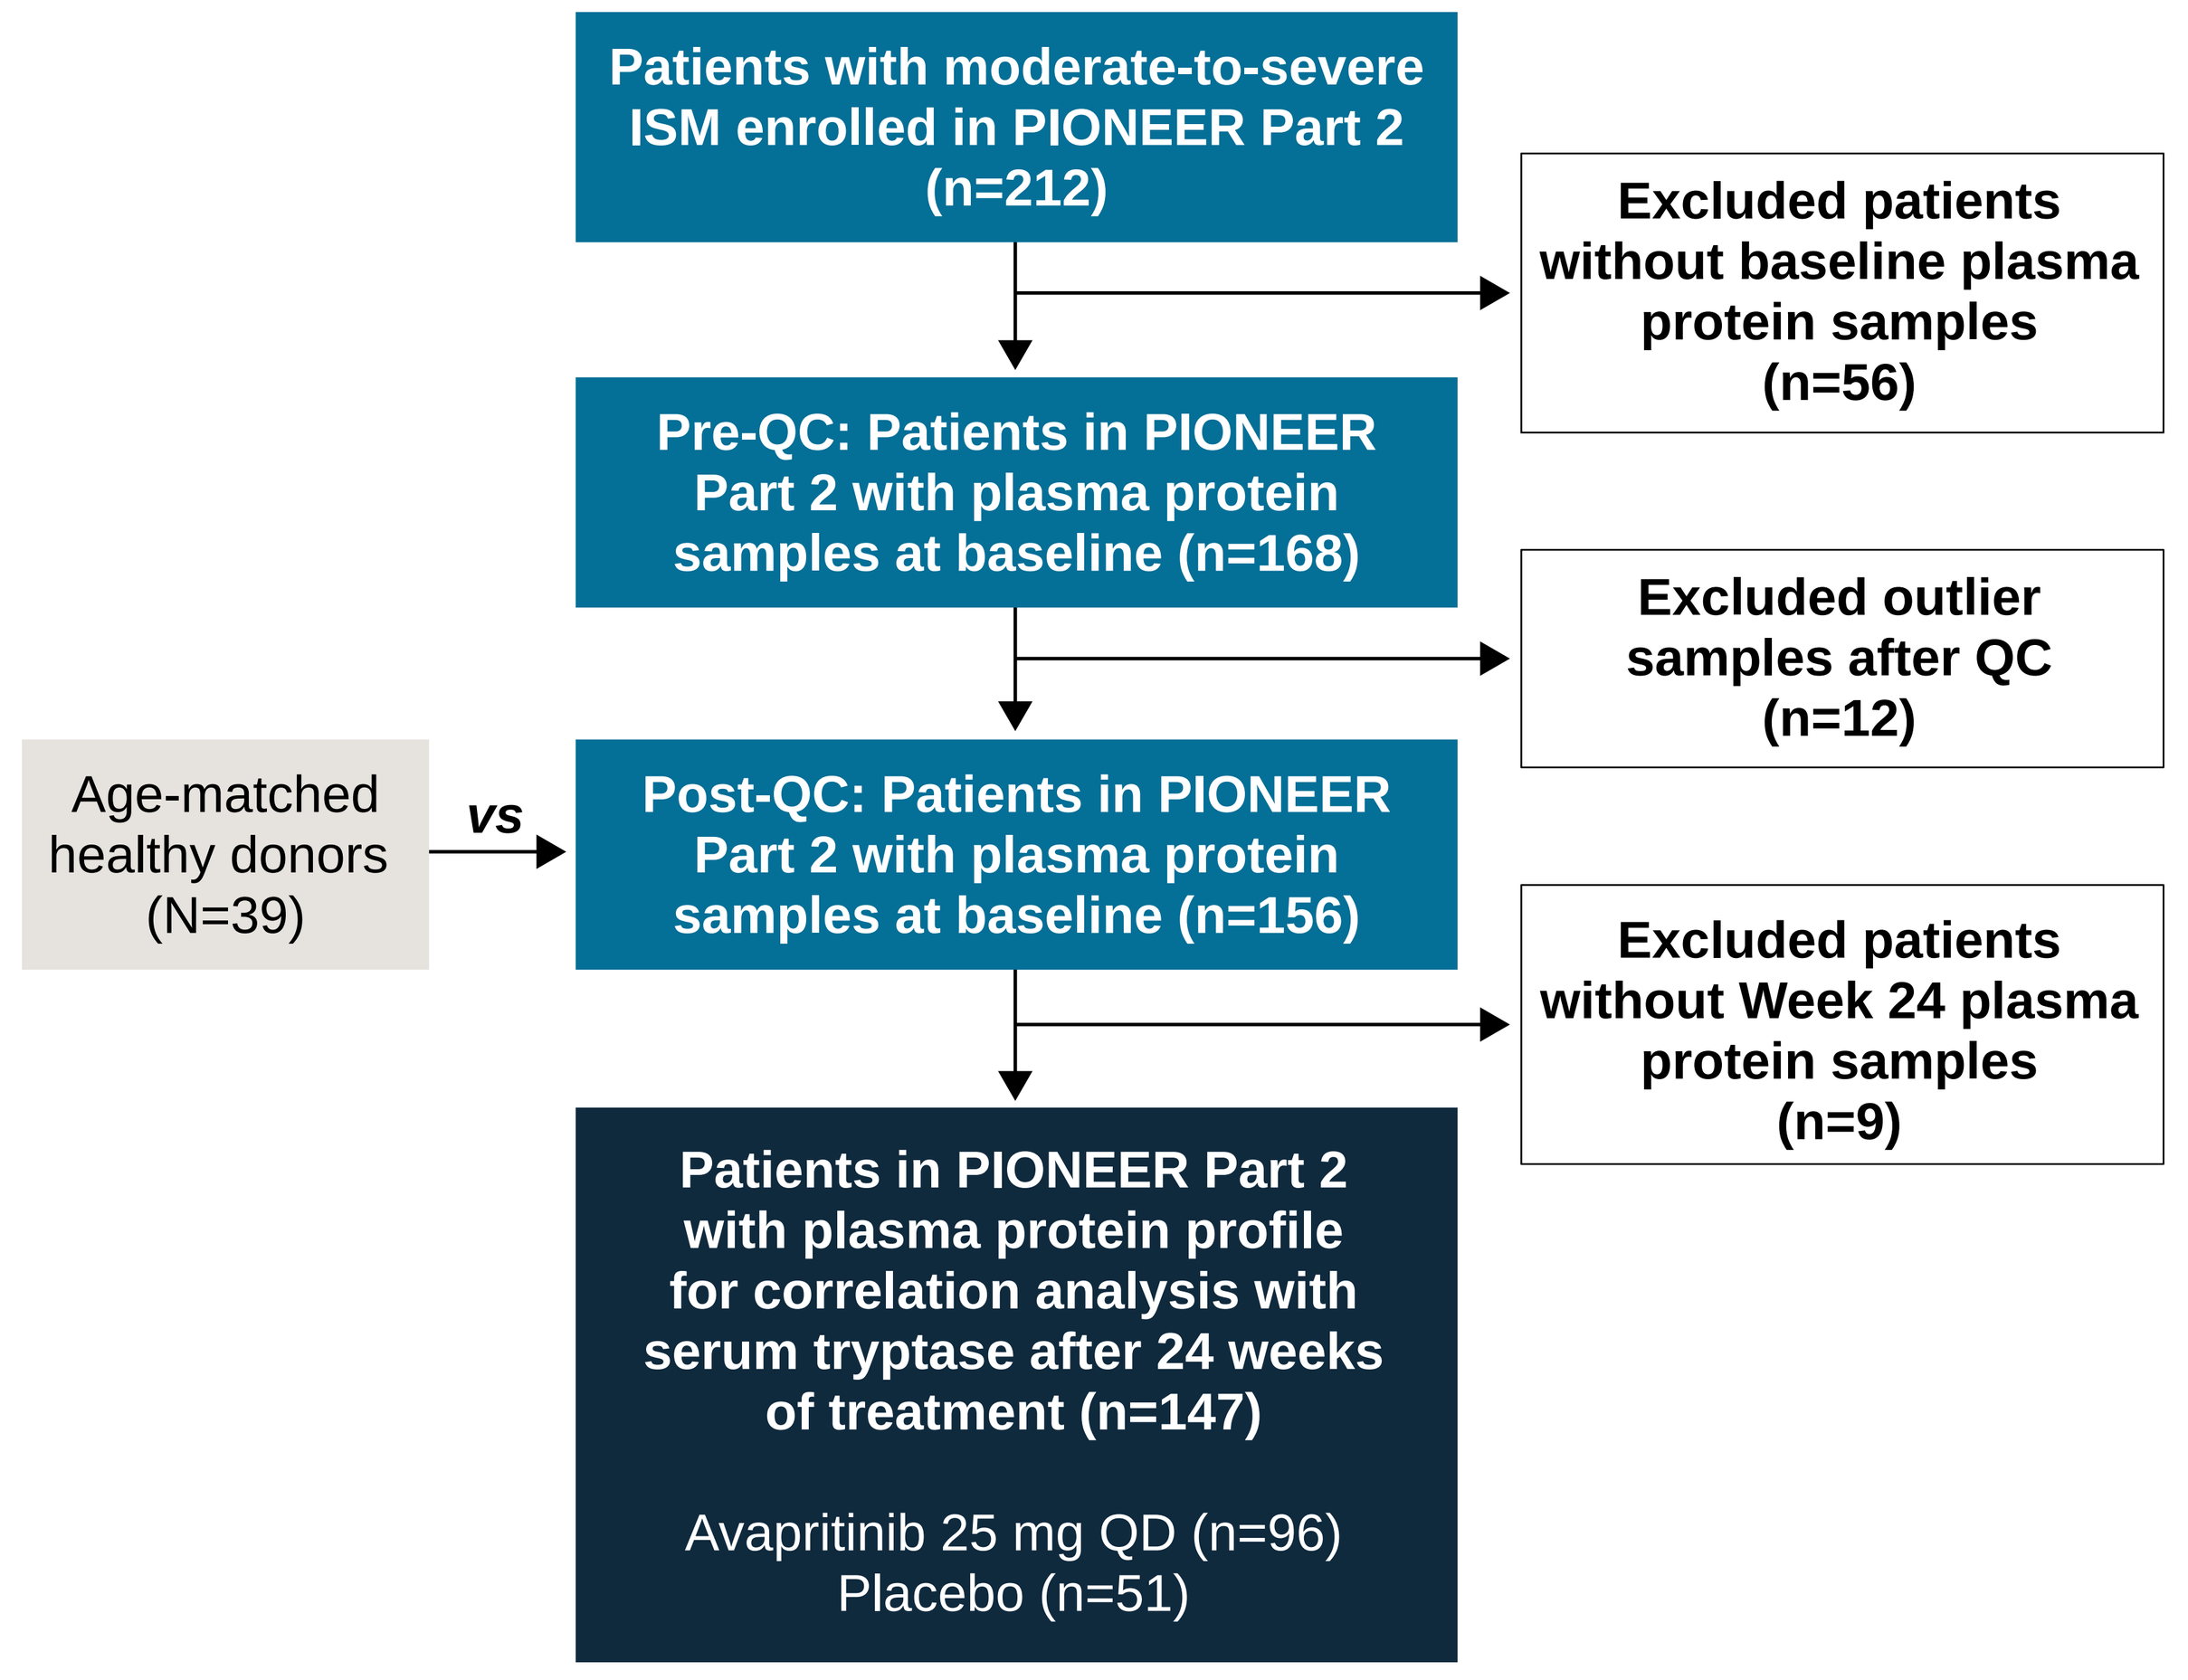

Patients with moderate-to-severe ISM enrolled in PIONEER Part 2 (n=212)
Excluded patients without baseline plasma protein samples (n=56)
Pre-QC: Patients in PIONEER Part 2 with plasma protein samples at baseline (n=168)
Excluded outlier samples after QC (n=12)
Age-matched healthy donors (N=39)
vs
Post-QC: Patients in PIONEER Part 2 with plasma protein samples at baseline (n=156)
Excluded patients without Week 24 plasma protein samples (n=9)
Patients in PIONEER Part 2 with plasma protein profile for correlation analysis with serum tryptase after 24 weeks of treatment (n=147)
Avapritinib 25 mg QD (n=96)
Placebo (n=51)

ISM, indolent systemic mastocytosis; QC, quality control; QD, once daily.

- Plasma samples from age-matched healthy donors (n=39) were assessed as controls and compared with baseline plasma protein samples from patients in PIONEER (n=156)
- Plasma protein measurements below the lower limit of quantification (LLOQ) were imputed as LLOQ values in the analysis
- Spearman's correlation coefficient (r) was determined to assess for an association between plasma proteins at baseline
- For baseline plasma protein comparison between healthy donors and patients with ISM, a t-test was performed for each protein and the resulting P-values were adjusted for multiple comparisons using the Benjamini-Hochberg method, false discovery rate (FDR)
 - An FDR of <0.05 was considered significant

Results

- Baseline demographics for the pooled population can be seen in **Table 1**
- At baseline, 156/363 proteins were significantly different in patients with ISM *versus* healthy donors (FDR <0.05) (**Figure 2**)

Table 1. Baseline demographics and disease characteristics

	PIONEER patients who received either avapritinib 25 mg QD or placebo (n=156)
Age (years), median (range)	52 (22–79)
Female, n (%)	119 (76)
ISM symptom burden	
Baseline ISM-SAF TSS, mean (SD)	51.7 (19.9)
MC burden	
Median (range) serum tryptase (central), ng/mL	44.3 (3.6–501.6)
Median (range) BM MCs, %	7.0 (1.0–70.0)
Median (range) *KIT* D816V VAF in peripheral blood, %	0.42 (undetectable–36.7)

BM, bone marrow; ISM-SAF, Indolent Systemic Mastocytosis Symptom Assessment Form (© 2018 Blueprint Medicines Corporation); MC, mast cell; SD, standard deviation; TSS, total symptom score; VAF, variant allele frequency.

Figure 2. Comparison of plasma proteins present in PIONEER patients with ISM *vs* healthy donors

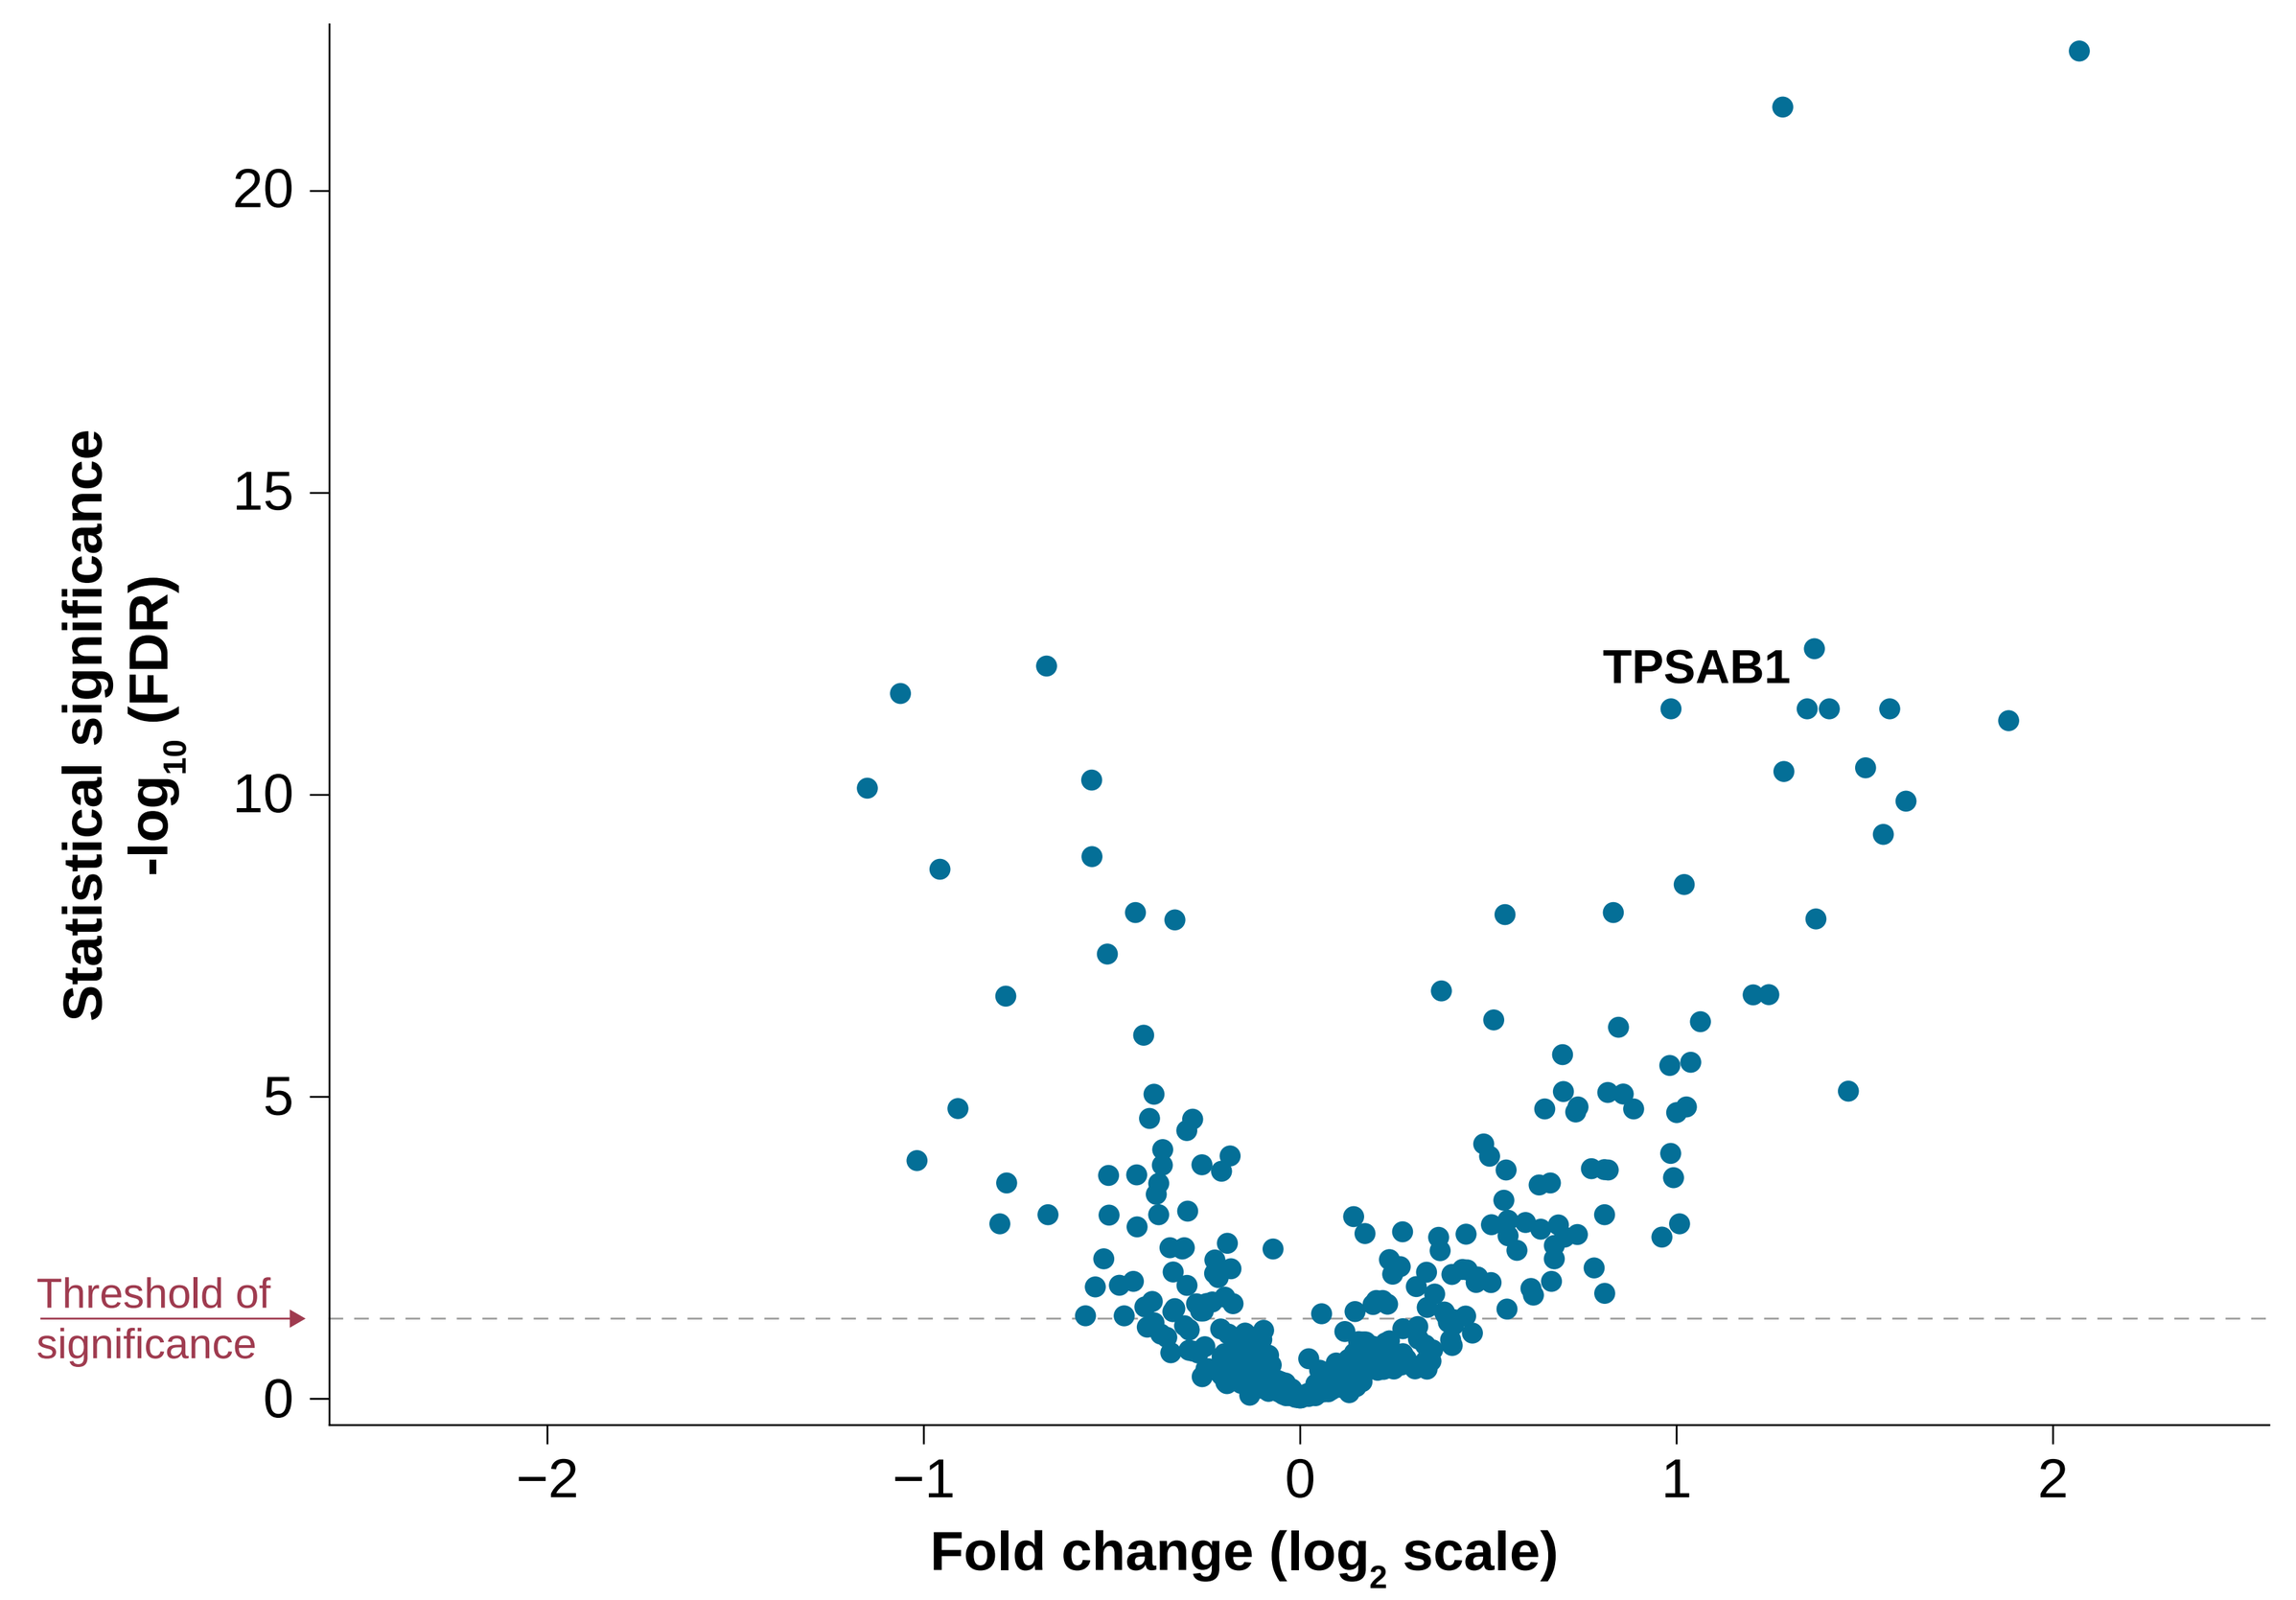

Statistical significance -log10 (FDR)
Threshold of significance
TPSAB1
Fold change (log2 scale)

FDR, false discovery rate.

- Tryptase measured by the Olink® panel moderately correlated (r=0.52) to clinically measured serum tryptase as a positive control
- The levels of many plasma proteins also correlated with clinically measured baseline serum tryptase levels, including (r-value): MILR1 (0.74), CCL23 (0.62), CD48 (0.51), SIGLEC10 (0.43), IL-5RA (0.40), CD4 (0.30), and IL-13 (0.23) (**Figure 3**)

Figure 3. Correlation between baseline plasma protein levels and clinically measured baseline tryptase levels in patients with ISM enrolled in PIONEER

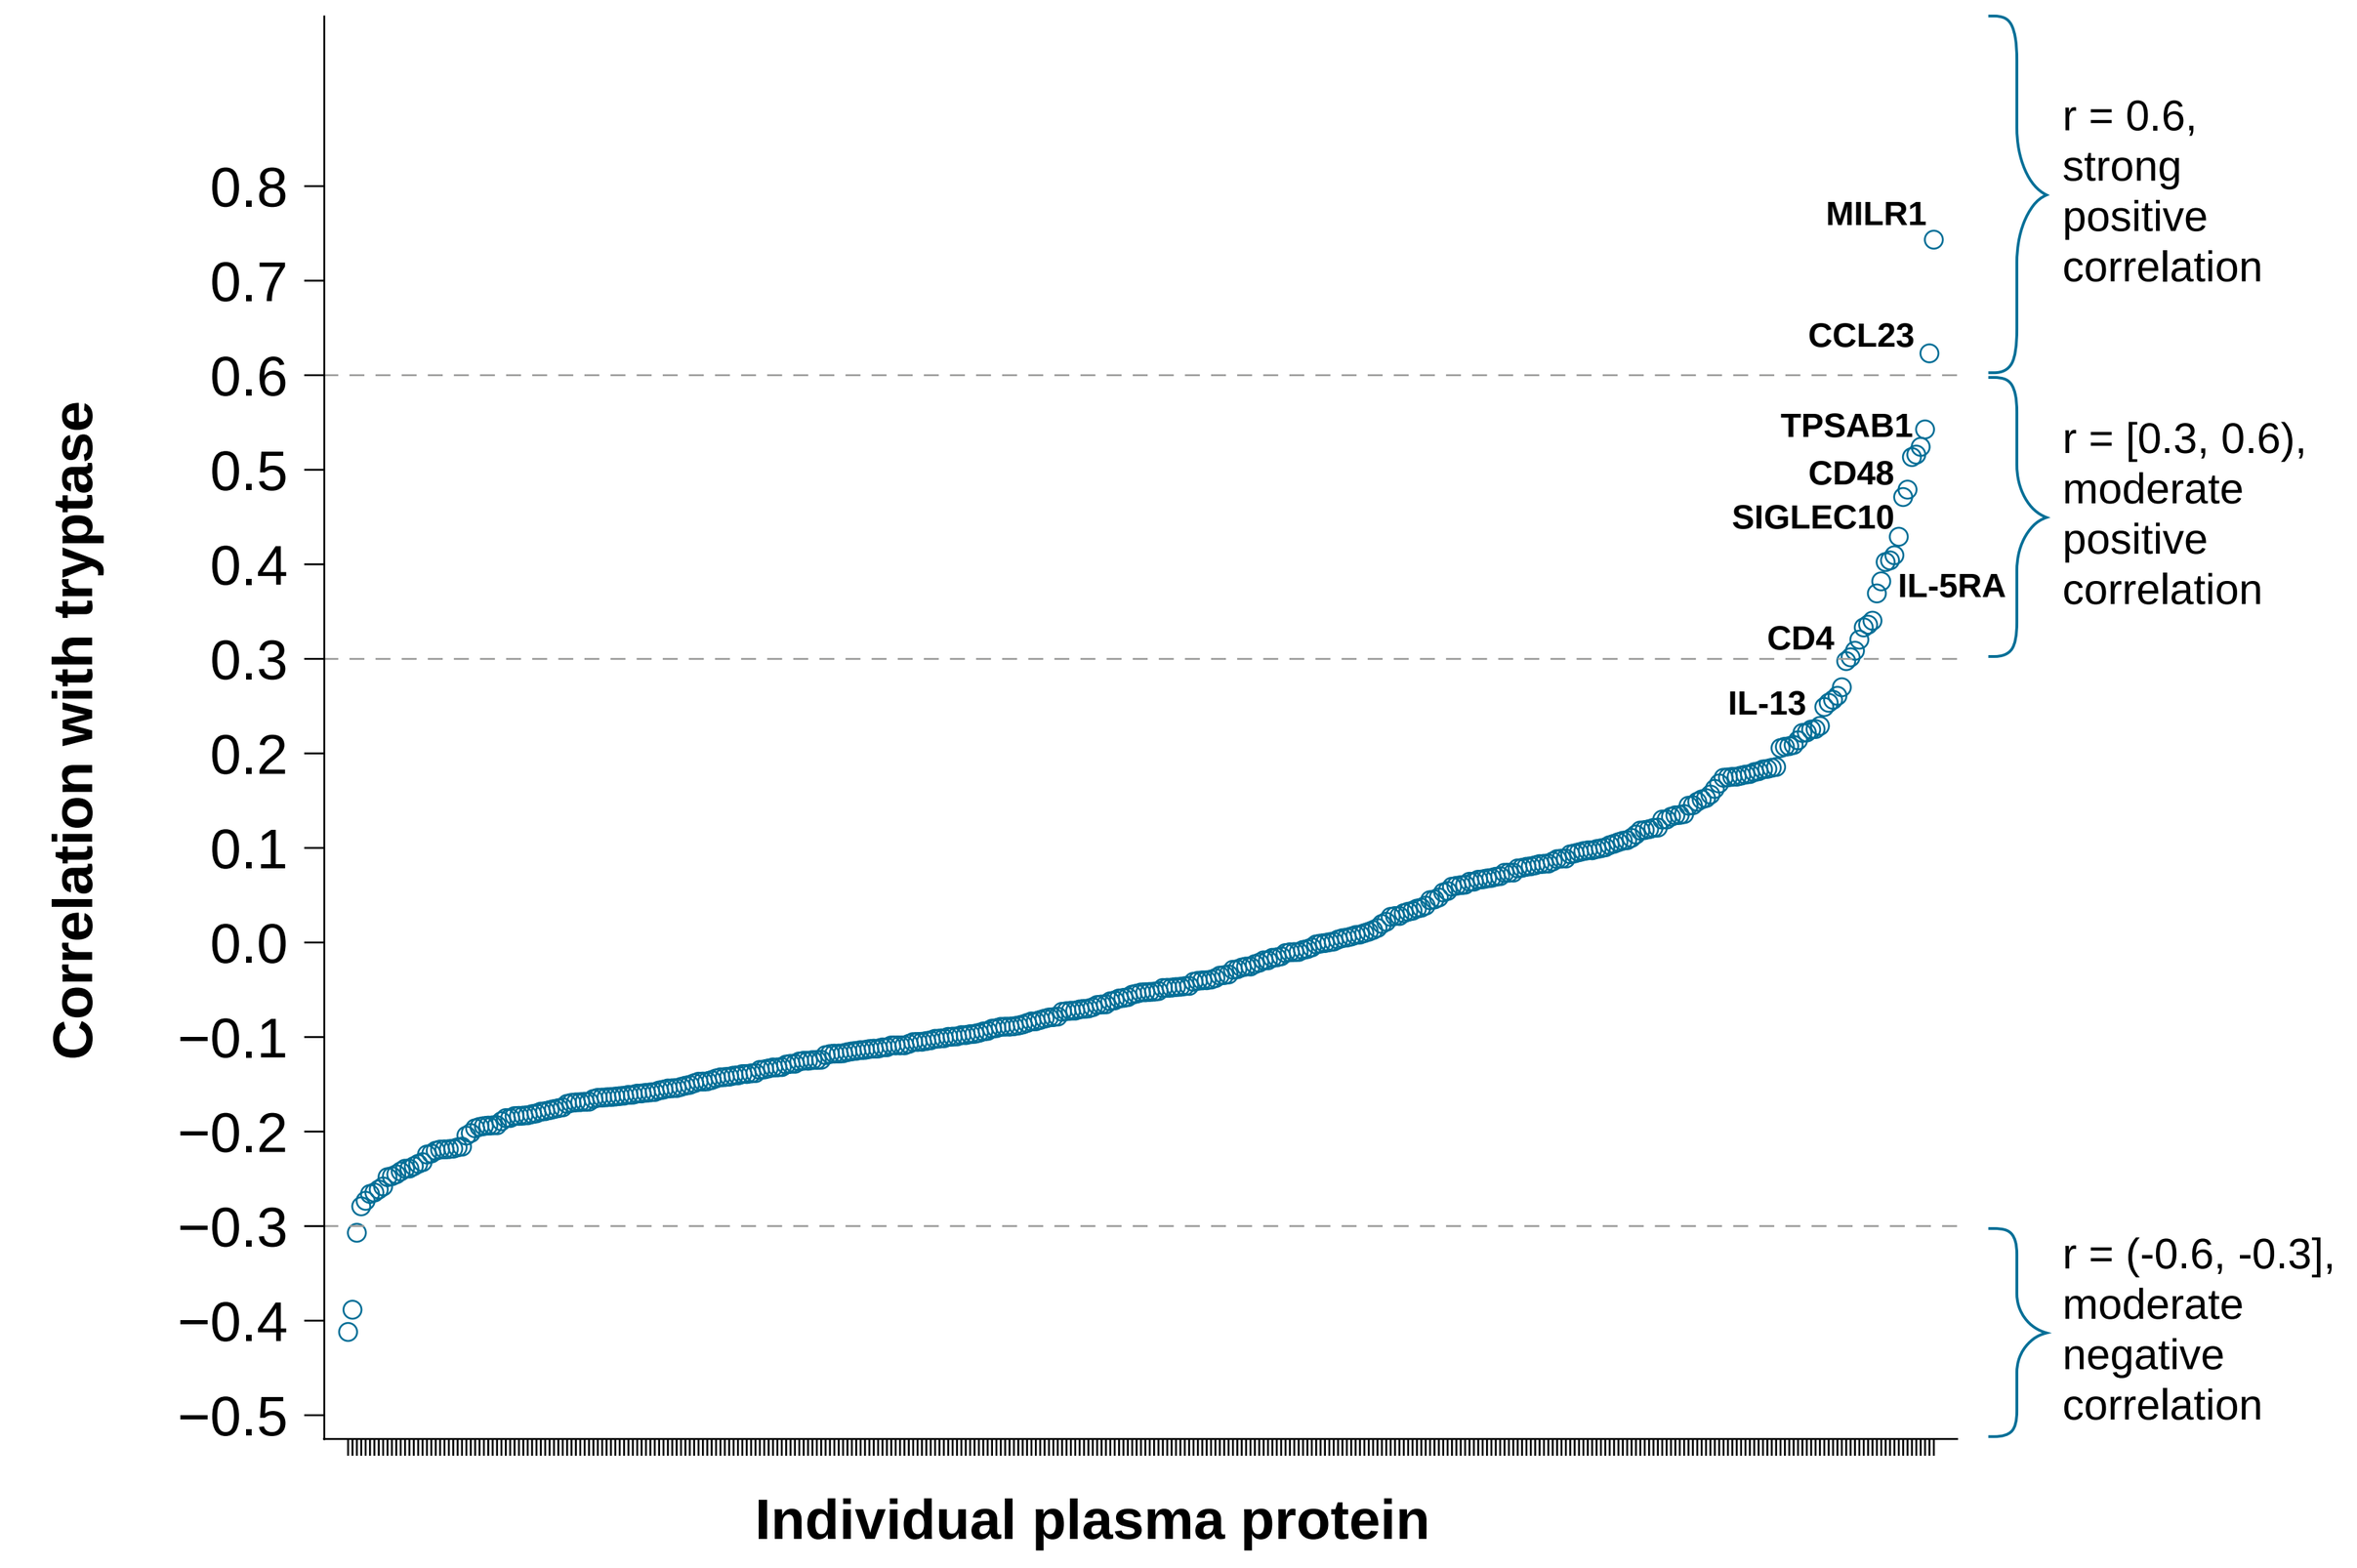

Correlation with tryptase
MILR1
CCL23
TPSAB1
CD48
SIGLEC10
IL-5RA
CD4
IL-13
r = 0.6, strong positive correlation
r = [0.3, 0.6), moderate positive correlation
r = (-0.6, -0.3], moderate negative correlation
Individual plasma protein

- Changes in serum tryptase levels with treatment were investigated, and were revealed to be significantly correlated with changes in MILR1, CCL23, CD48, and SIGLEC10 (FDR <0.05) (**Figure 4**)

Figure 4. Association of change in plasma protein levels with change in tryptase levels after 24 weeks in PIONEER

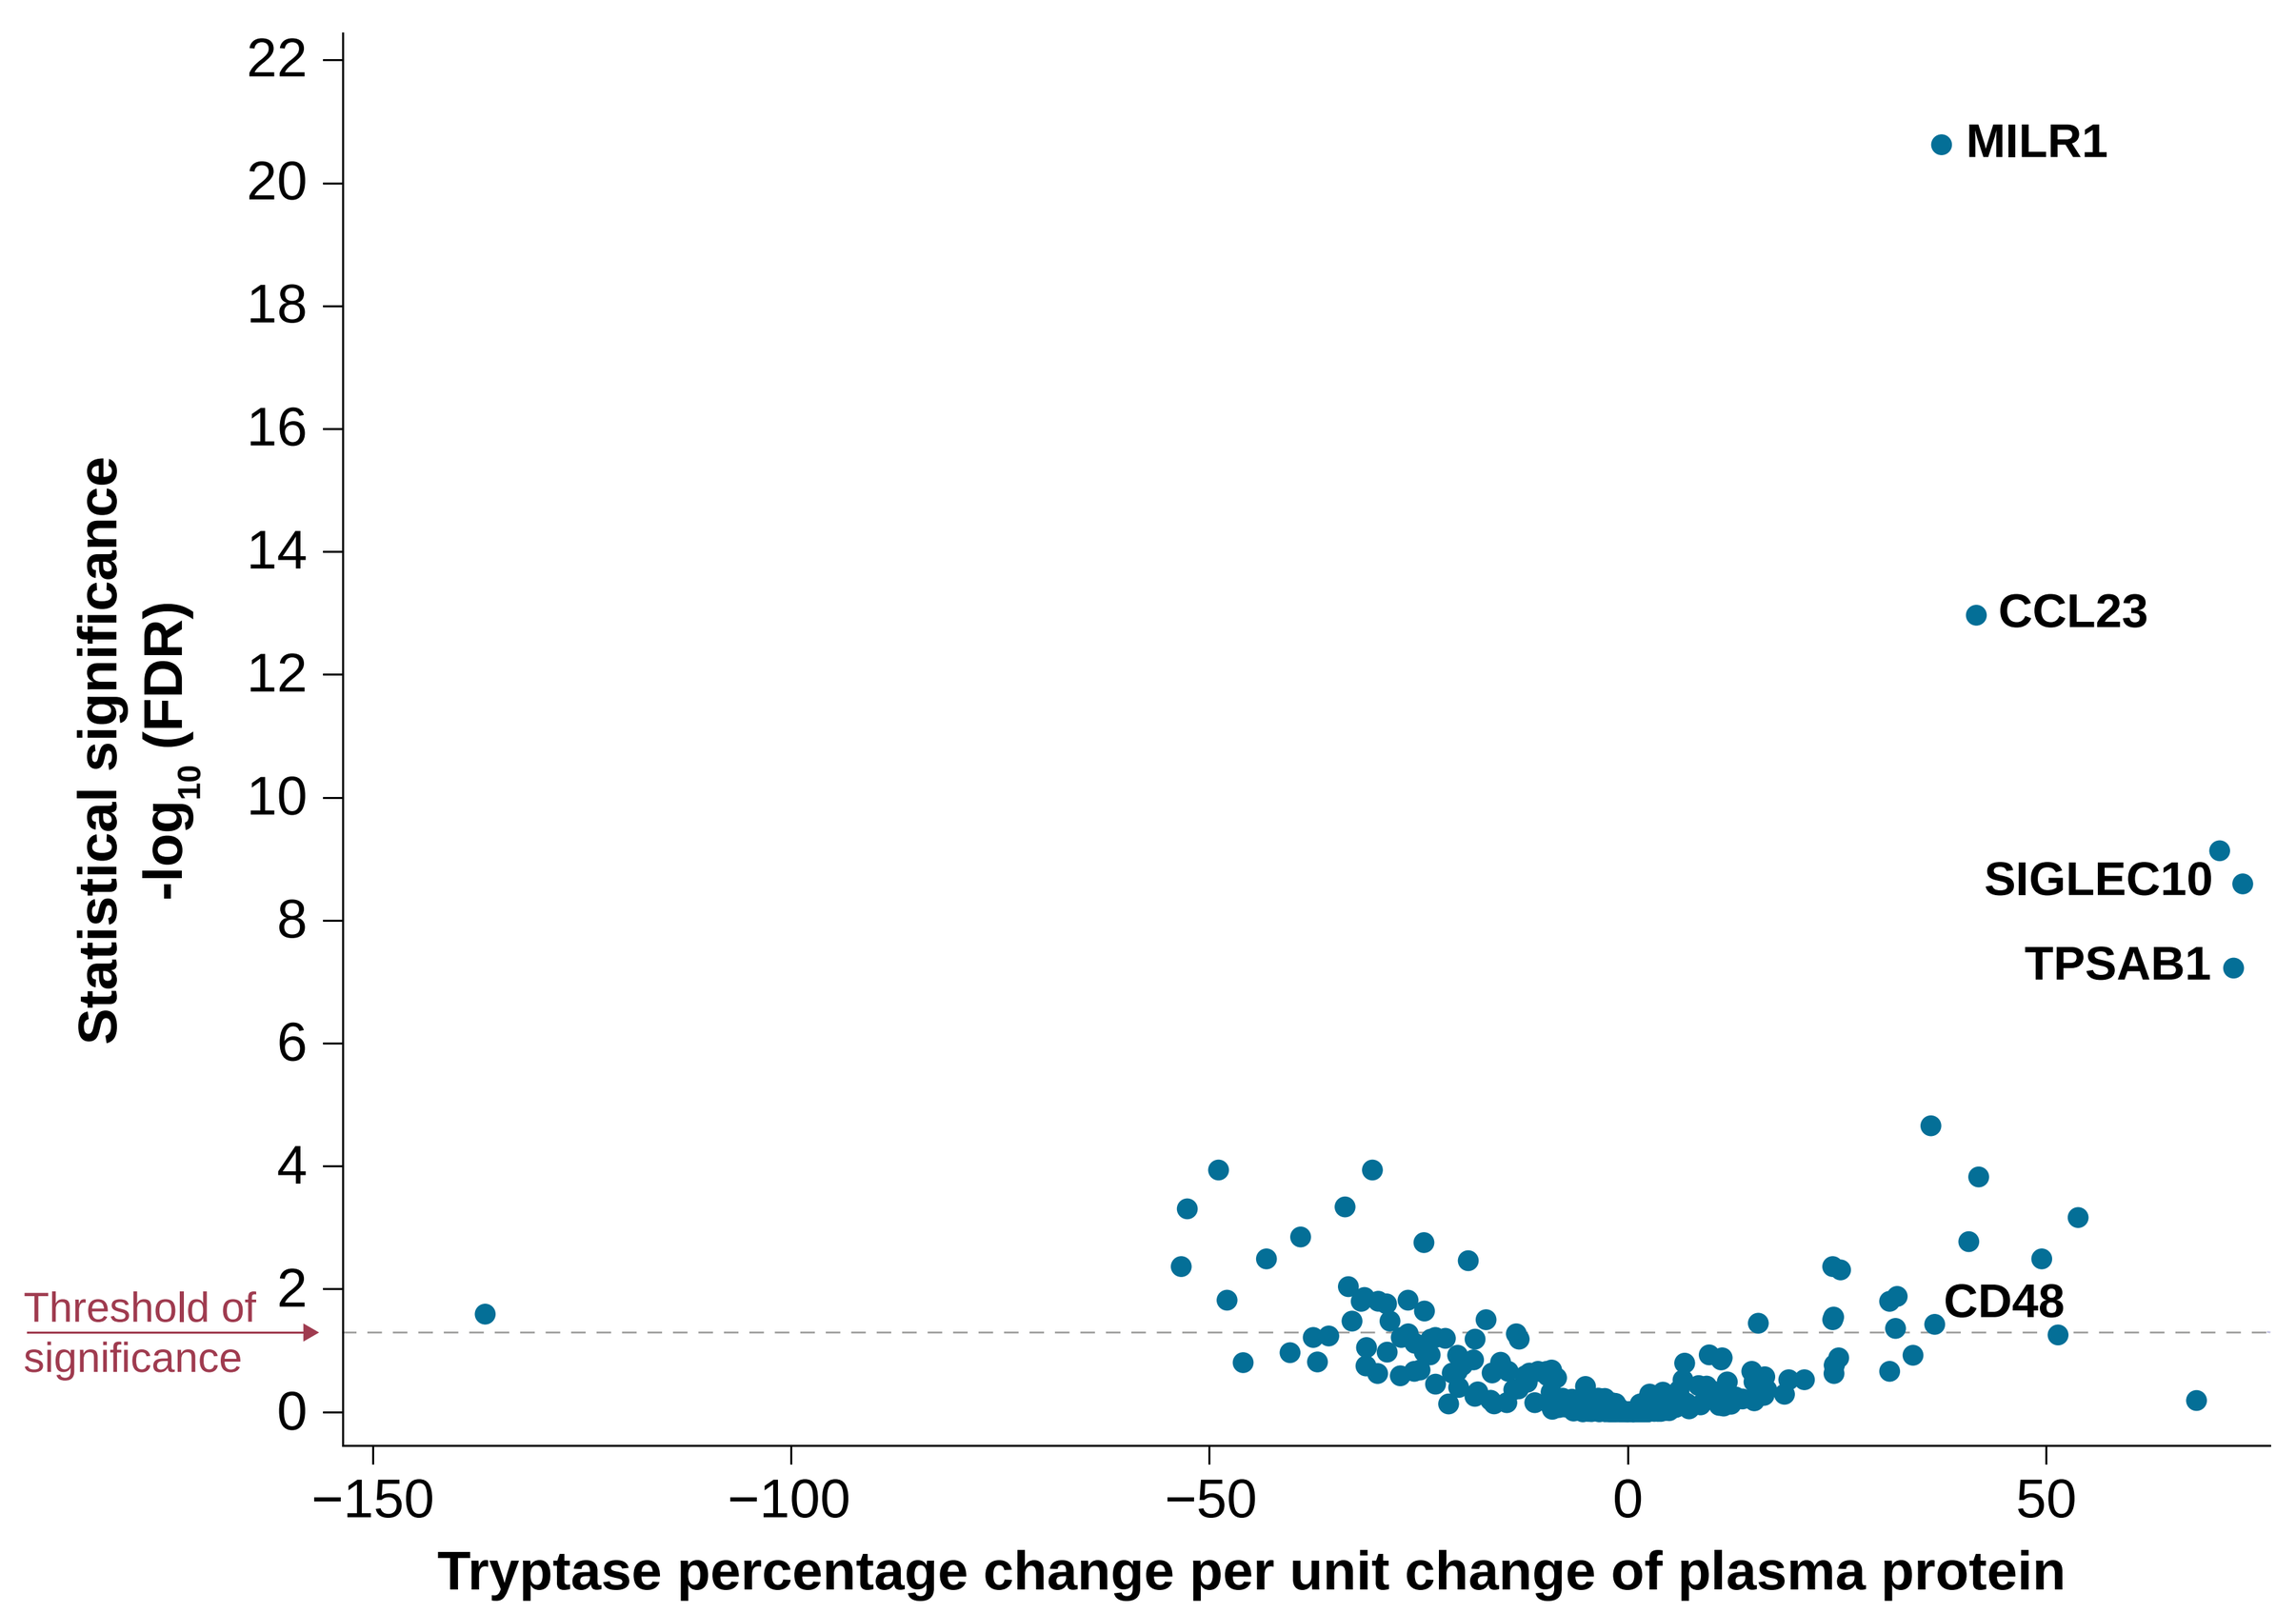

Statistical significance -log10 (FDR)
MILR1
CCL23
SIGLEC10
TPSAB1
CD48
Threshold of significance
Tryptase percentage change per unit change of plasma protein

Results

- Many of the proteins identified with expression patterns similar to tryptase are involved in regulating or modulating the immune system
 - MILR1 regulates the activation of MCs[17]
 - CCL23 is produced by MCs and its levels correlate with the severity of disease, with the highest levels seen in patients with advanced SM[18,19]
 - IL-13 is a key inflammatory cytokine in the type 2 inflammatory response, which has previously been implicated in ISM pathogenesis[20,21]
 - IL-5RA and associated signaling pathways play a key role in the MC – eosinophil interaction and is activated in patients with ISM[22]

Conclusions

- Comprehensive plasma proteome profiling is a useful method for understanding the large-scale changes that occur in the circulating inflammatory proteome in patients with ISM
- These findings highlight aberrant inflammatory signaling pathways in ISM, raising the possibility of combination therapy with KIT D816V inhibitors to address both the MC component and the inflammatory component of this disease
- Future work will also aim to identify whether inflammatory proteome analysis can identify biomarkers that correlate with symptom severity

References

1. Bergström A et al. *Acta Oncol*. 2024;63:44–50; 2. Kristensen T et al. *Am J Hematol*. 2014;89:493–498; 3. Ungerstedt J et al. *Cancers*. 2014;14:3942; 4. Garcia-Montero AC et al. *Blood*. 2006;108:2366–2372; 5. Pardanani A. *Am J Hematol*. 2023;98:1097–1116; 6. Mesa RA et al. *Cancer*. 2022;128:3691–3699; 7. Hermine O et al. *PLoS One*. 2008;3:e2266; 8. van Anrooij B et al. *Allergy*. 2016;71:1585–1593; 9. Akin C et al. *J Allergy Clin Immunol*. 2022;149:1912–1918; 10. Payne V et al. *Anaesthesia*. 2004;59:695–703; 11. Khoury JD et al. *Leukemia*. 2022;36:1703–1719; 12. Sánchez-Muñoz L et al. *Mod Path*. 2011;24:1157–1168; 13. Evans EK et al. Sci Transl Med. 2017;9:e1690; 14. Tashi T et al. Annual Meeting of the American Society of Hematology 2023, Oral #76; 15. Ayvakit® (avapritinib) Prescribing Information. Cambridge, MA: Blueprint Medicines Corporation; 2024; 16. Ayvakyt (avapritinib) Summary of Product Characteristics. Cambridge, MA; Blueprint Medicines Corporation; 2024; 17. Nanatsue K et al. *J Hum Genet.* 2014;59:480–483; 18. Mayado A et al. *Leukemia*. 2016;30:124–130; 19. Söderlund S et al. *J Allergy Clin Immunol*. 2023;152:205–213; 20. Hermans MAW et al. *J Allergy Clin Immunol*. 2022;150:146–156.e10; 21. Bao K et al. *Cytokine*. 2015;75:25–37; 22. Lefèvre G et al. *J Allergy Clin Immunol*. 2024;154:1523–1533.

Acknowledgements

Writing and editorial support under the direction of the authors was provided by Matt Nicolas, MSc, Danielle Scheunemann, MS, and Sarah Christopher, PhD, all of Paragon (a division of Prime, Knutsford, UK), funded by Blueprint Medicines Corporation.

Poster available for download at:

Disclosures

Dr Sabato's institution has received research funding from Blueprint Medicines Corporation for clinical trials. He received payment or honoraria for lectures, presentations, speakers' bureaus, manuscript writing, or educational events from Blueprint Medicines Corporation, Novartis, and Cogent. He participated in a Data Safety Monitoring Board or advisory board for Blueprint Medicines Corporation and Cogent.

For all author disclosures, please contact medinfo@blueprintmedicines.com.

Presented at the 2025 AAAAI/WAO Joint Congress; February 28–March 3, 2025, San Diego, CA. Please contact medinfo@blueprintmedicines.com for permission to reprint and/or distribute.