Avapritinib Impacts the Plasma Inflammatory Proteome in Patients With Indolent Systemic Mastocytosis (ISM)

Poster Number 517

Cem Akin,[1] Cristina Bulai Livideanu,[2] Ivan Alvarez-Twose,[3] Hussein A. Abbas,[4] Min Chen,[5] Guang Yang,[5] Rachel L. Erlich,[5] Benjamin Lampson,[5] Mariana Castells[6]

[1]University of Michigan, Ann Arbor, MI; [2]Hospital Center University De Toulouse, Toulouse, France; [3]Institute of Mastocytosis Studies of Castilla-La Mancha, Virgen del Valle Hospital, Toledo, Spain; [4]Department of Leukemia, Division of Cancer Medicine, MD Anderson Cancer Center, Houston, TX; [5]Blueprint Medicines Corporation, Cambridge, MA; [6]Mastocytosis Center, Brigham and Women's Hospital and Harvard Medical School, Boston, MA

Introduction

- The prevalence of systemic mastocytosis (SM) has been estimated at up to 1 in 5,000 people[1]
- Indolent systemic mastocytosis (ISM), the most common subtype of SM, is a clonal mast cell (MC) disease driven by the *KIT* D816V mutation in ~95% of patients[2–5]
- Patients with ISM often experience life-long debilitating symptoms across multiple organ systems[6–10]
 - These symptoms are likely due to a combination of direct MC organ infiltration, MC activation and mediator release, and inflammatory immune system alterations promoted by aberrant MCs[10]
- Avapritinib is a potent, oral inhibitor that selectively targets the KIT D816V mutation,[11] with approximately 30-fold selectivity for the mutant KIT protein over the wild-type KIT protein[12]
- In the PIONEER trial (NCT03731260) studying patients with moderate-to-severe ISM, avapritinib significantly reduced symptom burden and biomarkers of MC burden, compared with placebo[13]
 - Avapritinib has been approved in the USA and Europe for adult patients with ISM based on these findings[14,15]
- The clonal expansion and activation of MCs with consequent release of mediators results in inflammatory changes which impact tissues and cells to an extent that has not been explored
- To further explore this, we leveraged the large and well-characterized population of patients enrolled in PIONEER to conduct high-throughput profiling of the inflammatory plasma proteome

Methods

- The Olink® Explore 384 Inflammation panel (Uppsala, Sweden) measured 363 well-established plasma proteins in plasma samples from patients in PIONEER and healthy donors (**Figure 1**)
- Plasma samples from age-matched healthy donors (n=39) were assessed as controls and compared with baseline plasma protein samples from patients in PIONEER (N=156)
- Plasma protein measurements below the lower limit of quantification (LLOQ) were imputed as LLOQ values in the analysis
- For baseline plasma protein comparison between healthy donors and patients with ISM, a t-test was performed for each protein and the resulting P-values were adjusted for multiple comparisons using the Benjamini-Hochberg method, false discovery rate (FDR)
 - An FDR of <0.05 was considered significant

Figure 1. Study design

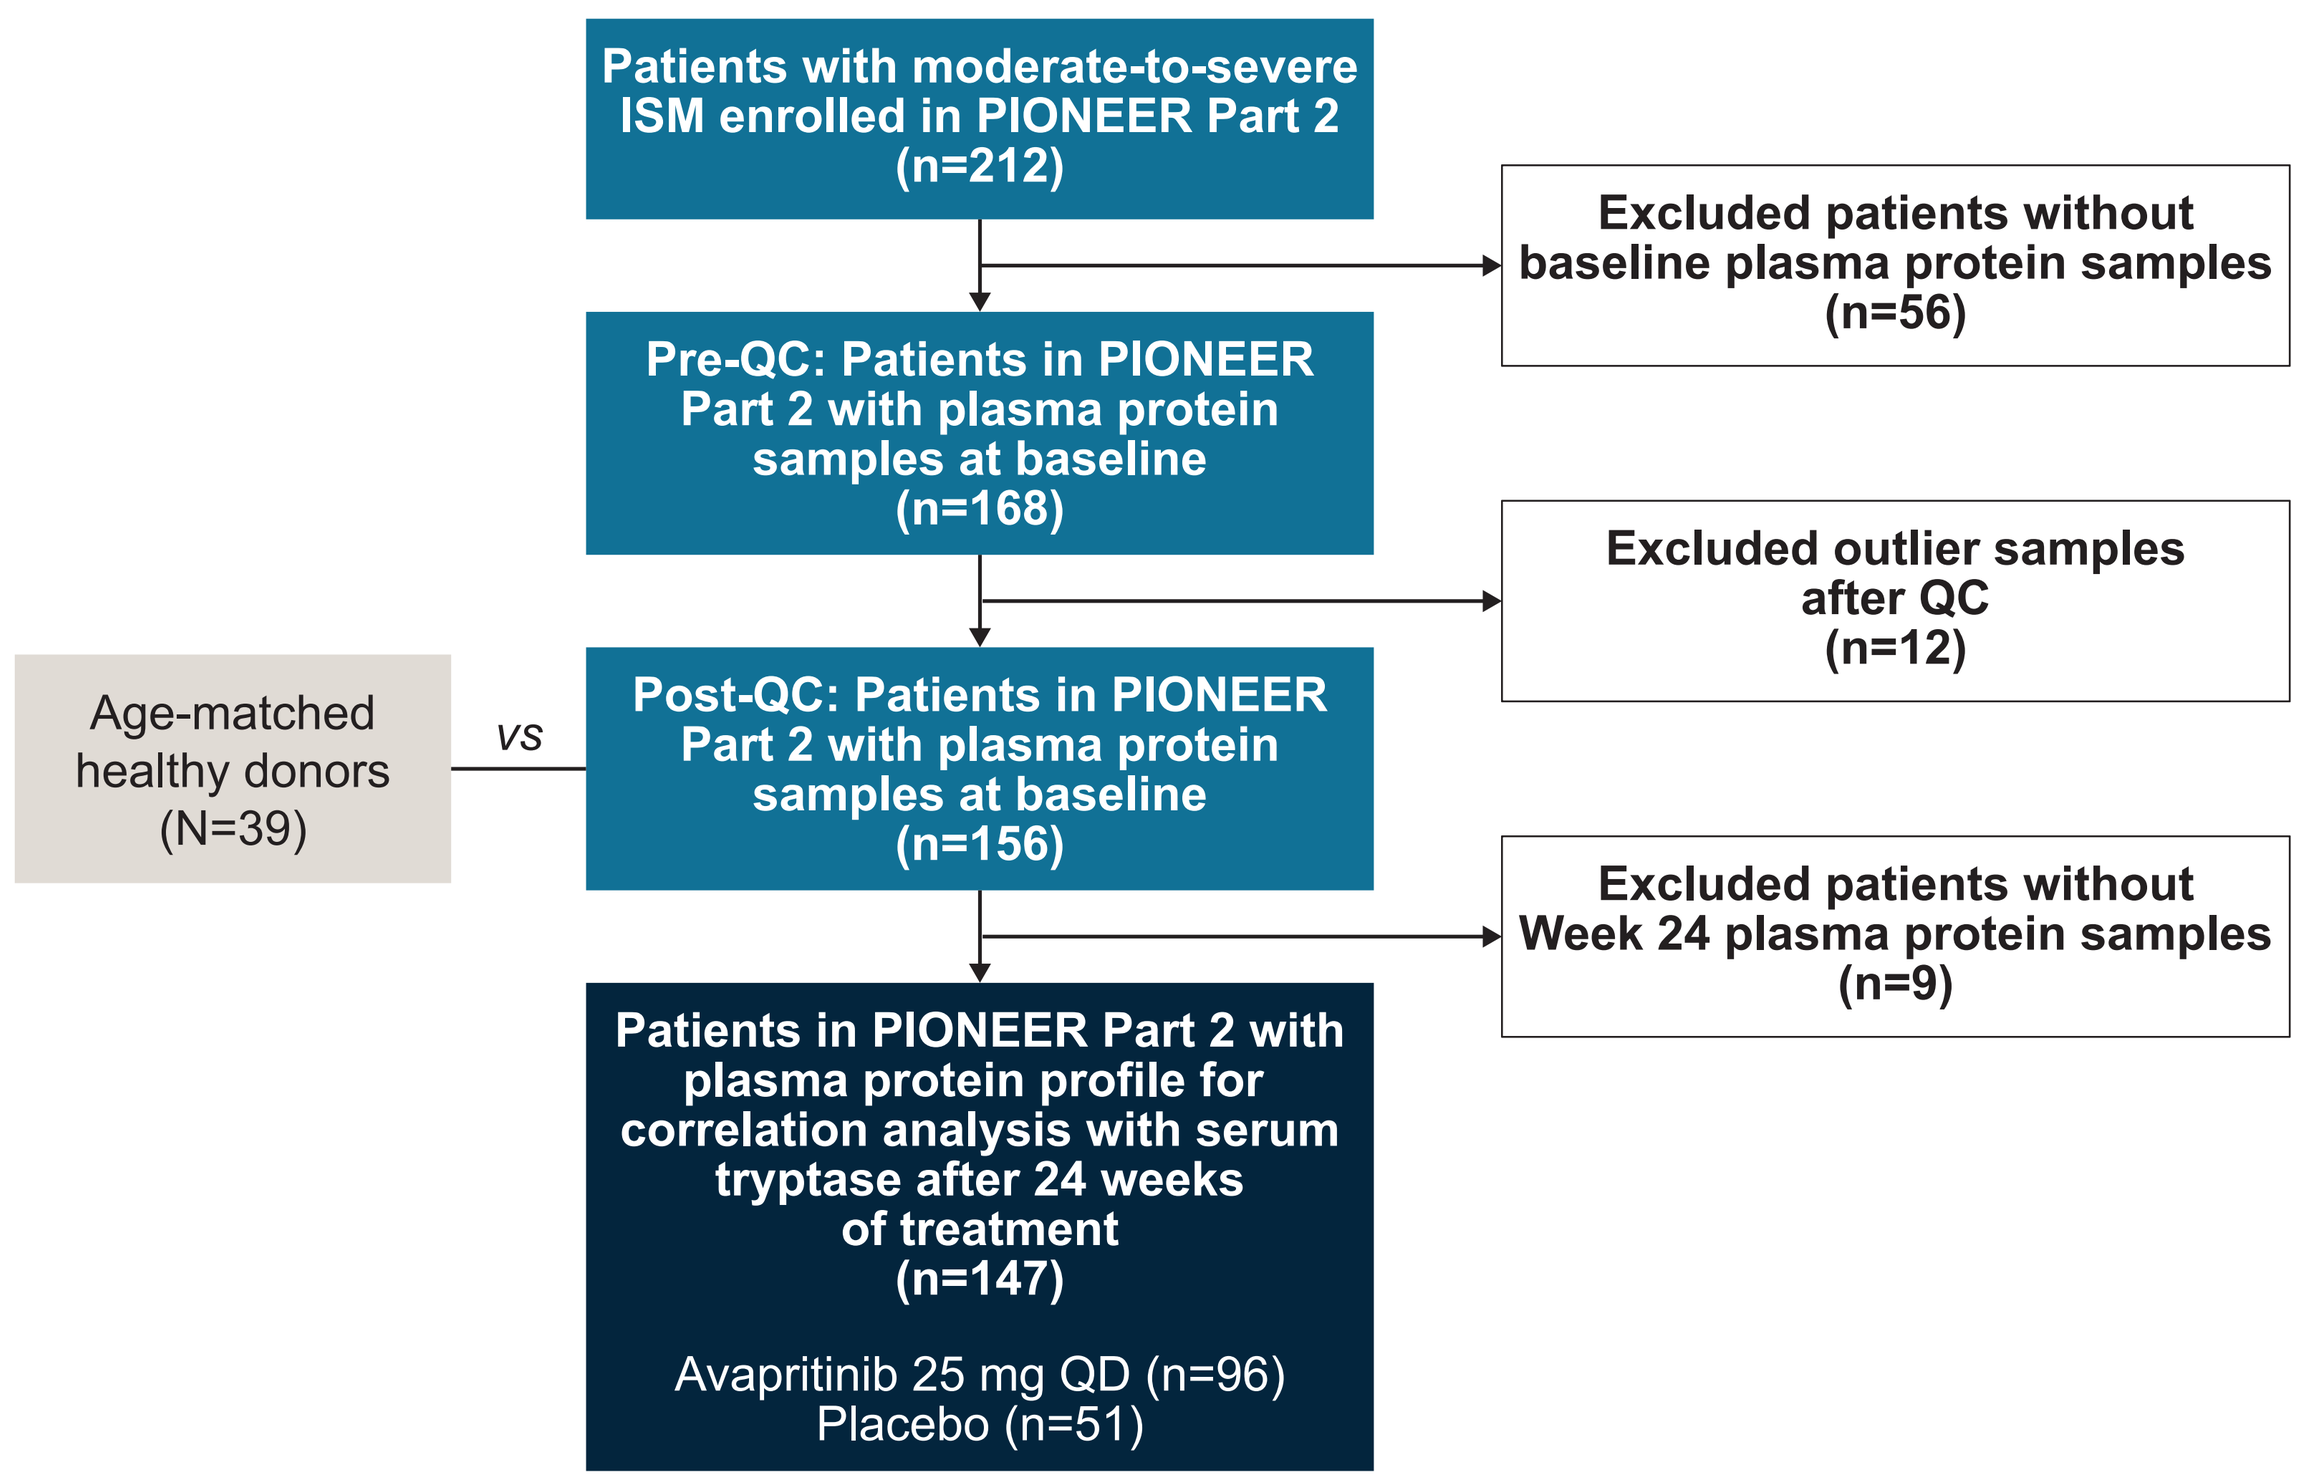

Patients with moderate-to-severe ISM enrolled in PIONEER Part 2 (n=212)
Excluded patients without baseline plasma protein samples (n=56)
Pre-QC: Patients in PIONEER Part 2 with plasma protein samples at baseline (n=168)
Excluded outlier samples after QC (n=12)
Age-matched healthy donors (N=39)
vs
Post-QC: Patients in PIONEER Part 2 with plasma protein samples at baseline (n=156)
Excluded patients without Week 24 plasma protein samples (n=9)
Patients in PIONEER Part 2 with plasma protein profile for correlation analysis with serum tryptase after 24 weeks of treatment (n=147)
Avapritinib 25 mg QD (n=96)
Placebo (n=51)

ISM, indolent systemic mastocytosis; QC, quality control; QD, once daily.

- For change from baseline to Week 24 after treatment, a paired t-test was performed for each protein
 - The raw P-values were adjusted for multiple comparisons using the same method above

Results

- Baseline demographics are presented in **Table 1**

Table 1. Baseline demographics and disease characteristics

	PIONEER patients who received either avapritinib 25 mg QD or placebo and had suitable samples for plasma protein analysis (n=156)
Age (years), median (range)	52 (22–79)
Female, n (%)	119 (76)
ISM symptom burden	
Baseline ISM-SAF TSS, mean (SD)	51.7 (19.9)
MC burden	
Median (range) baseline serum tryptase, ng/mL	44.3 (3.6–501.6)
Median (range) baseline BM MCs, %	7.0 (1.0–70.0)
Median (range) baseline *KIT* D816V VAF in peripheral blood, %	0.42 (undetectable–36.7)

BM, bone marrow; ISM-SAF, Indolent Systemic Mastocytosis Symptom Assessment Form (© 2018 Blueprint Medicines Corporation); MC, mast cell; SD, standard deviation; TSS, total symptom score; VAF, variant allele frequency.

Figure 2. Comparison of baseline plasma proteins present in patients with ISM *vs* healthy donors

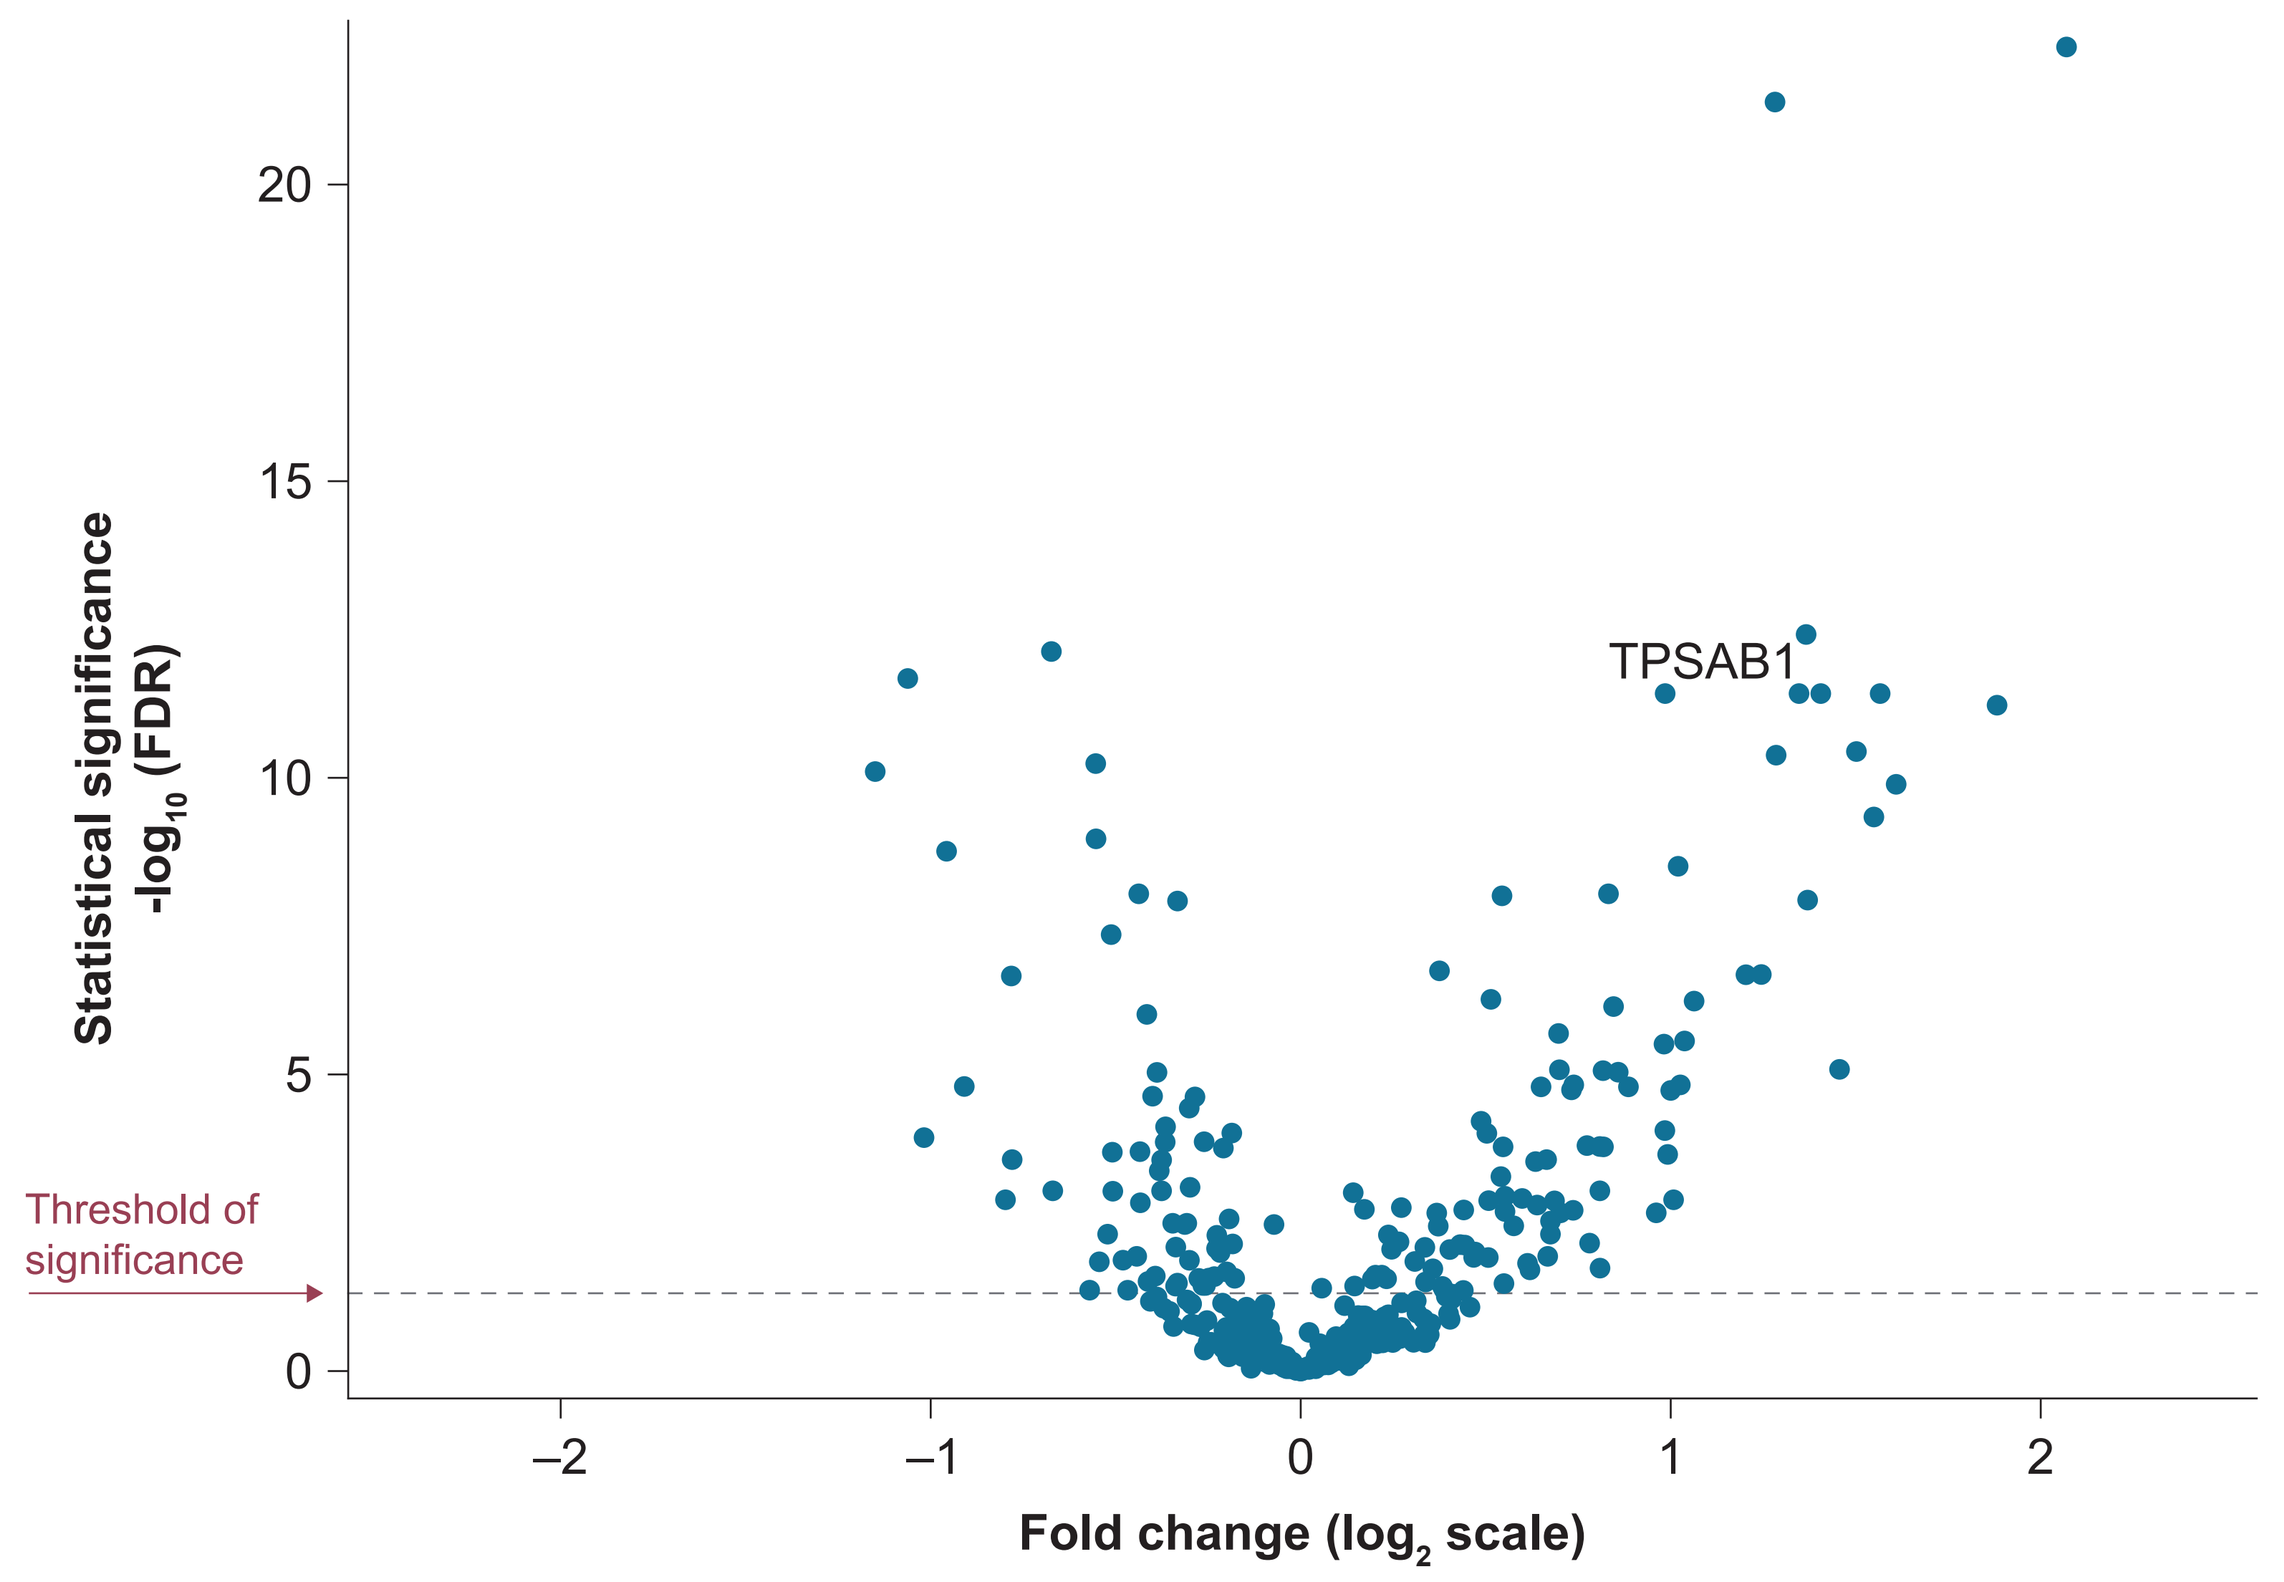

Statistical significance
$-\log_{10}$ (FDR)
20
15
10
5
0
TPSAB1
Threshold of significance
–2
–1
0
1
2
Fold change ($\log_2$ scale)

FDR, false discovery rate.

- At baseline, 156/363 proteins were significantly different in patients with ISM *versus* healthy donors (FDR <0.05) (**Figure 2**)
- After 24 weeks of avapritinib therapy, 76/363 proteins differed significantly from baseline (FDR <0.05) (**Figure 3**)

Figure 3. Plasma protein changes in patients in PIONEER treated with 24 weeks of avapritinib

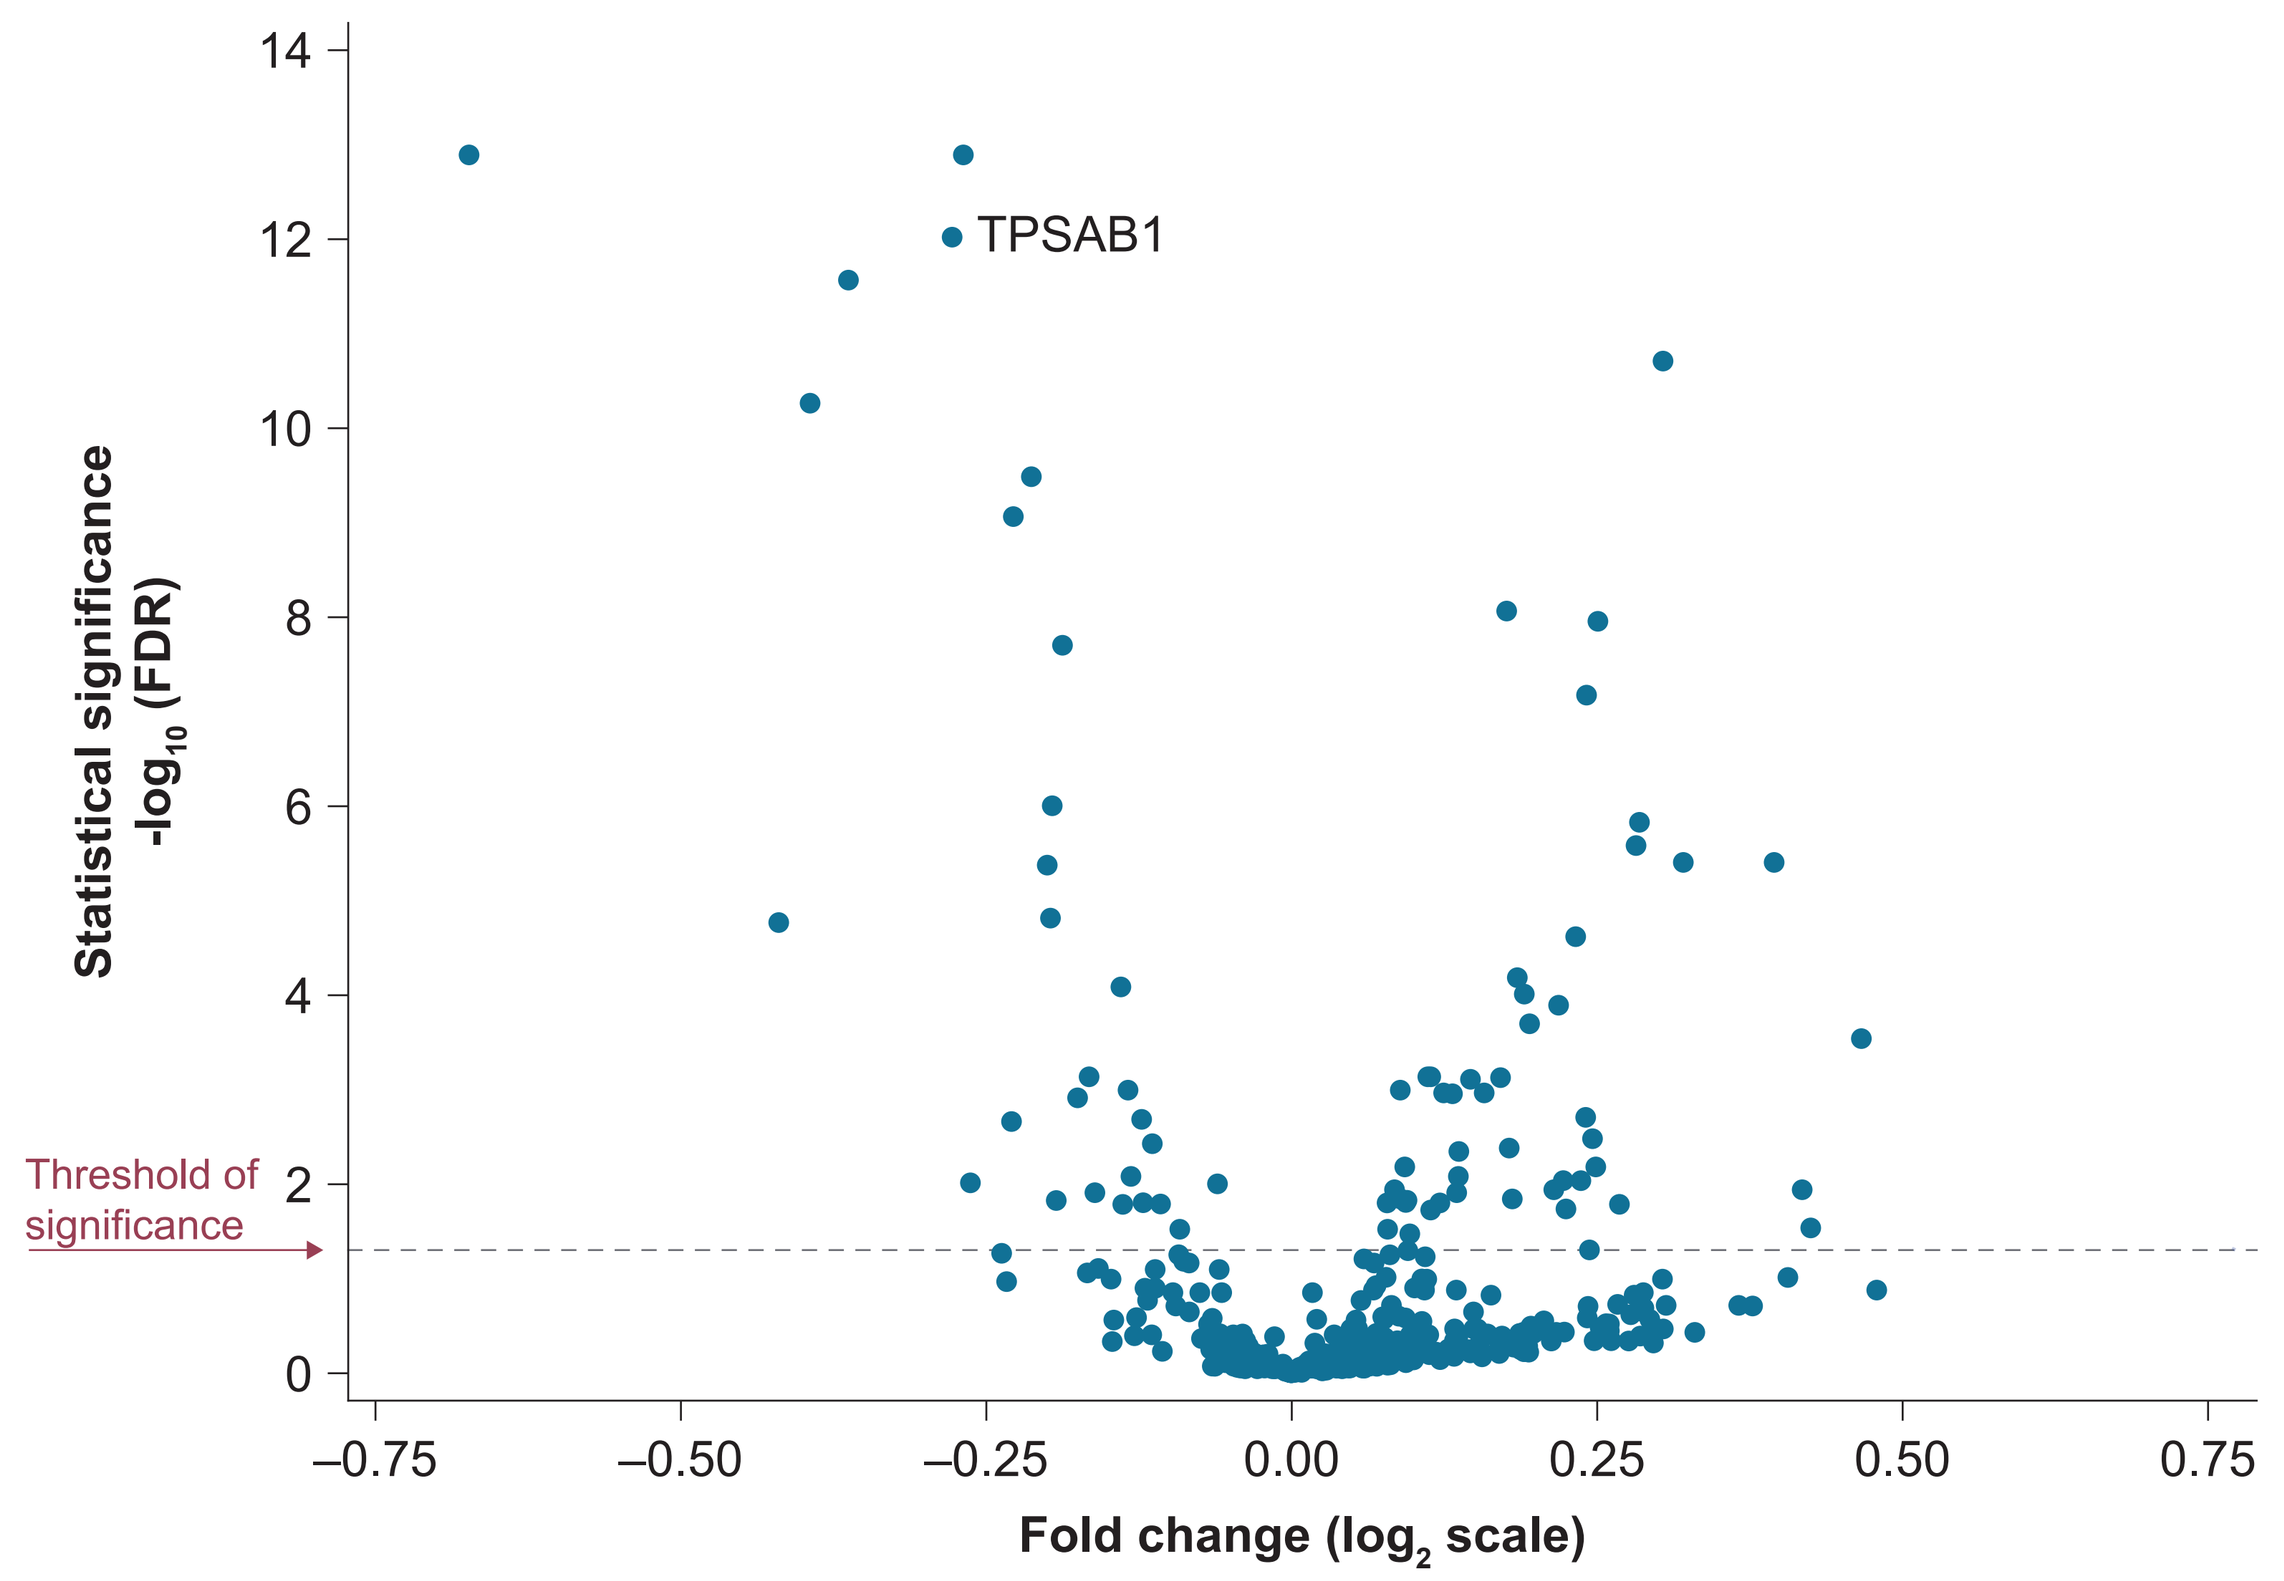

Statistical significance
$-\log_{10}$ (FDR)
14
12
10
8
6
4
2
0
TPSAB1
Threshold of significance
–0.75
–0.50
–0.25
0.00
0.25
0.50
0.75
Fold change ($\log_2$ scale)

Figure 4. Plasma protein changes in patients in PIONEER receiving 24 weeks of placebo

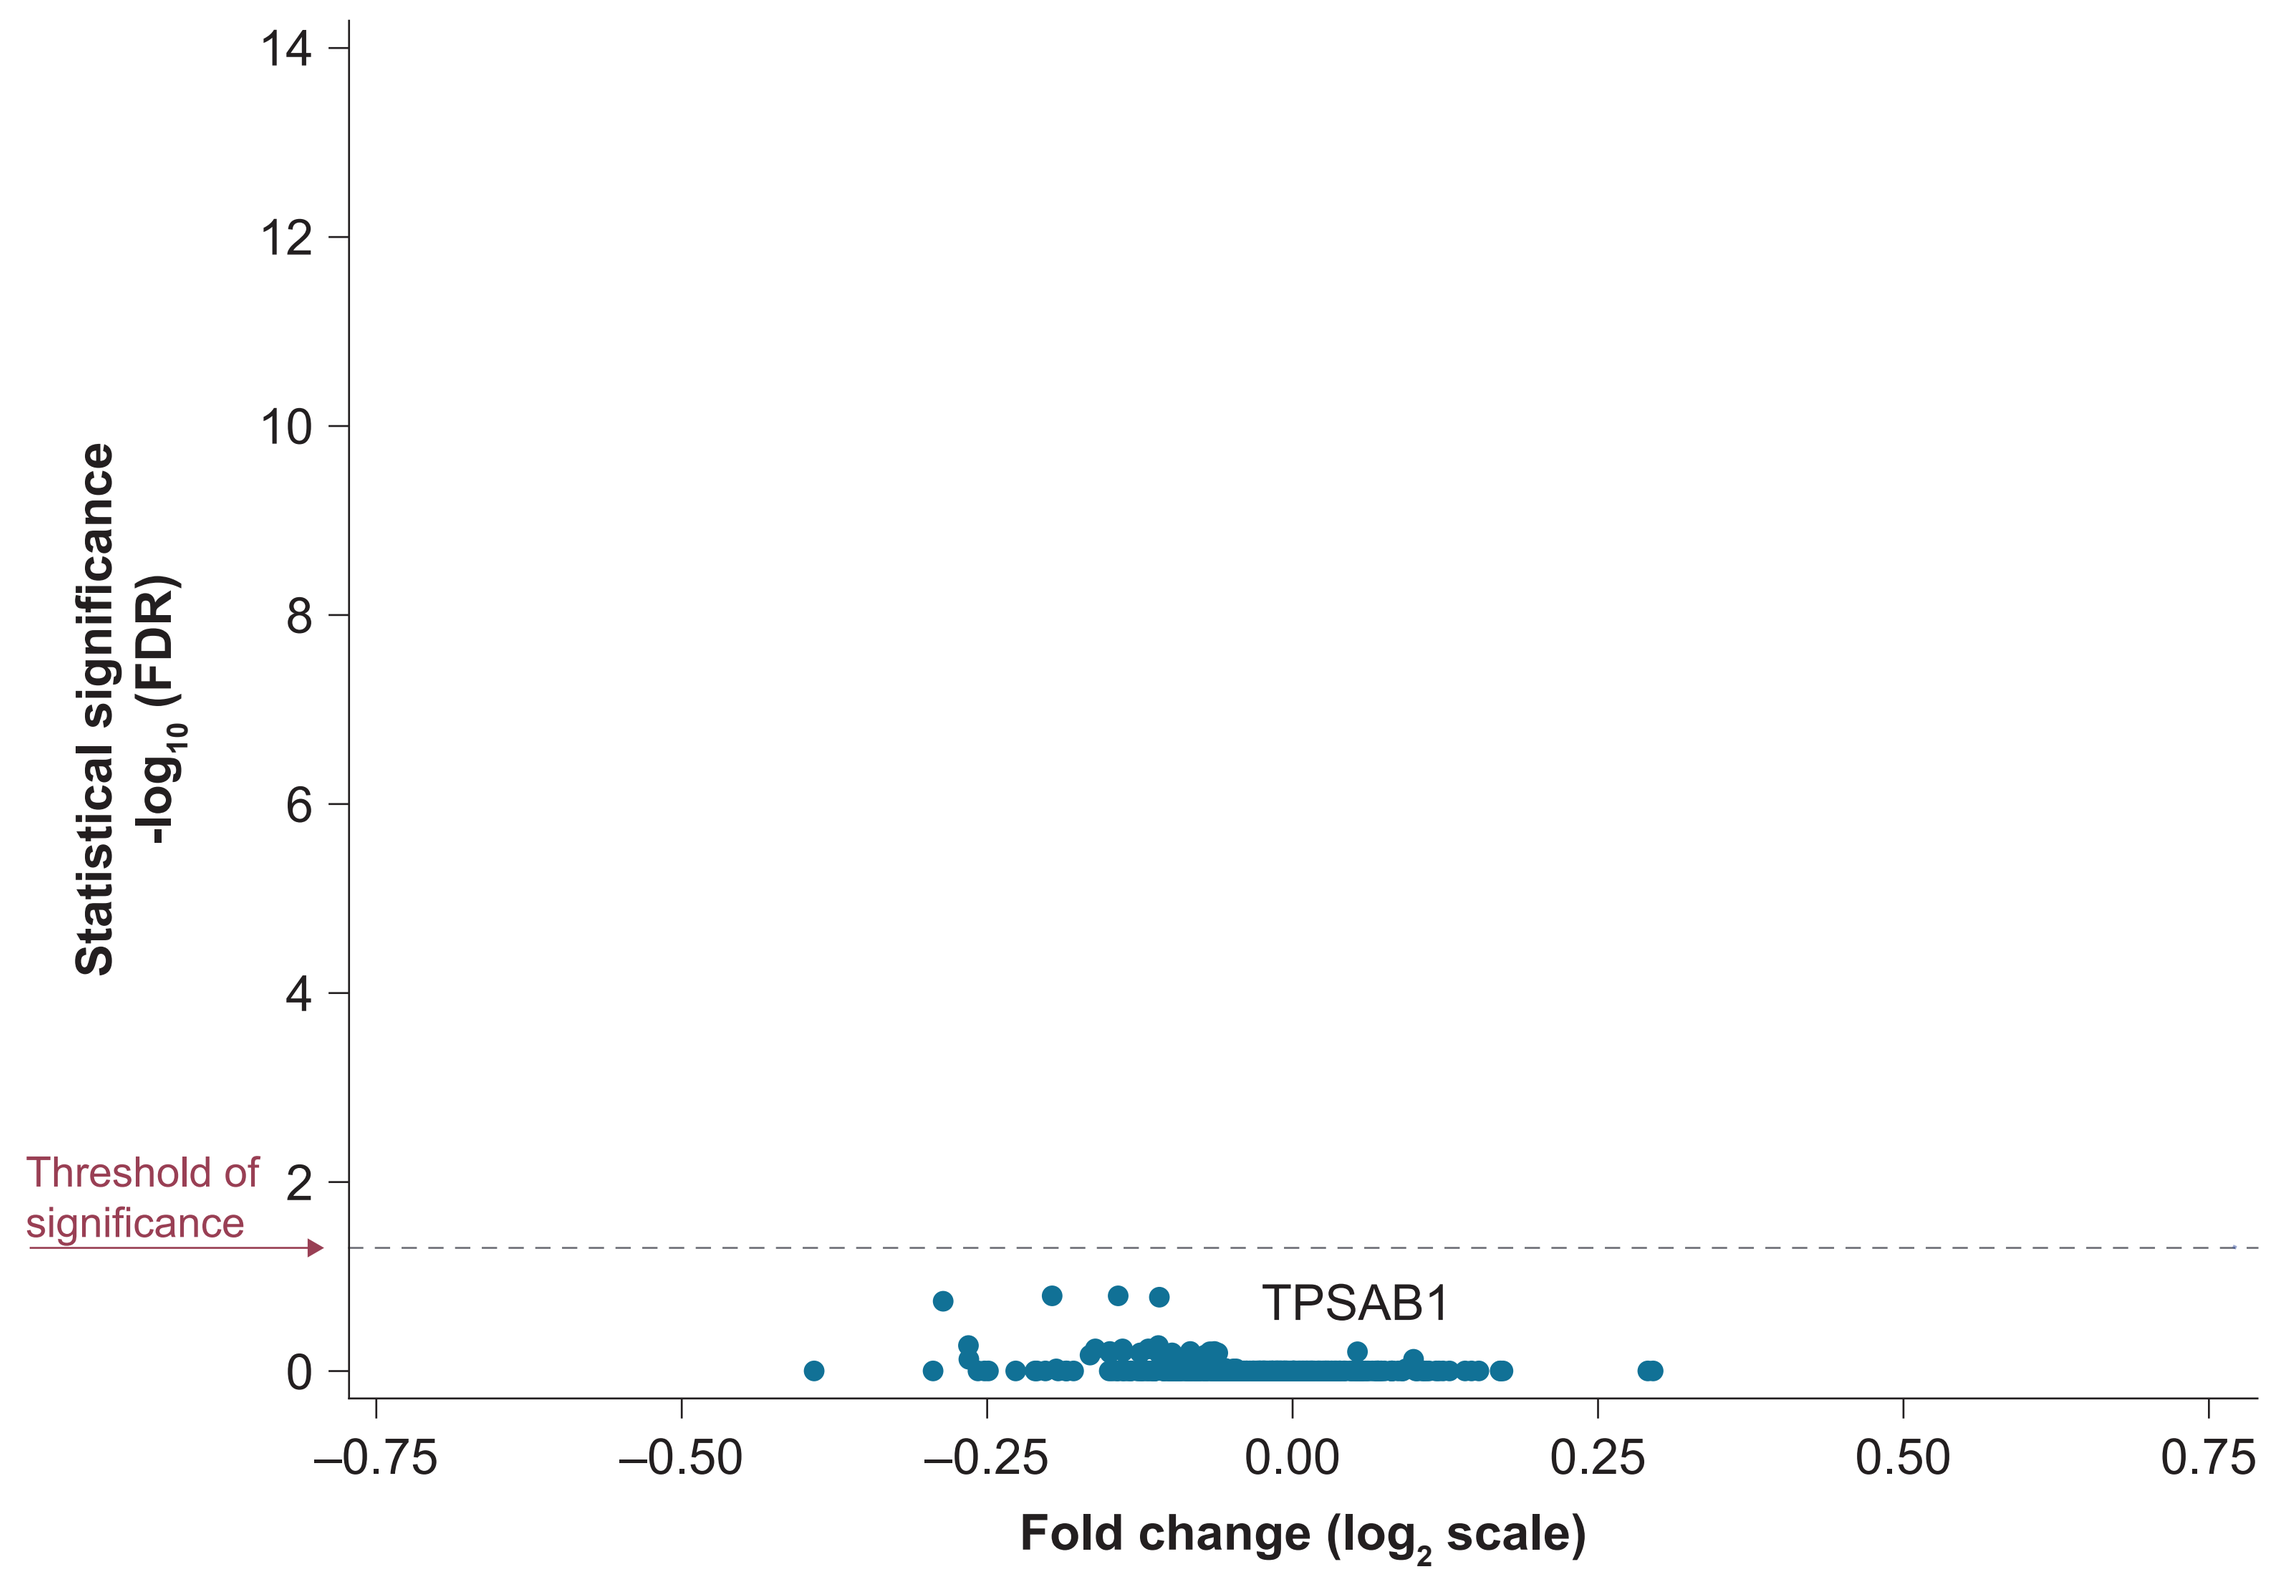

Statistical significance
$-\log_{10}$ (FDR)
14
12
10
8
6
4
2
0
Threshold of significance
TPSAB1
–0.75
–0.50
–0.25
0.00
0.25
0.50
0.75
Fold change ($\log_2$ scale)

- No proteins showed significant changes in placebo-treated patients (**Figure 4**)
- Out of the 156 proteins identified as ISM-specific, six significantly decreased with avapritinib therapy and 25 significantly increased (**Figure 5**)

Figure 5. Overlap between plasma proteins significantly different in patients with ISM *versus* plasma proteins significantly changed by avapritinib treatment for 24 weeks

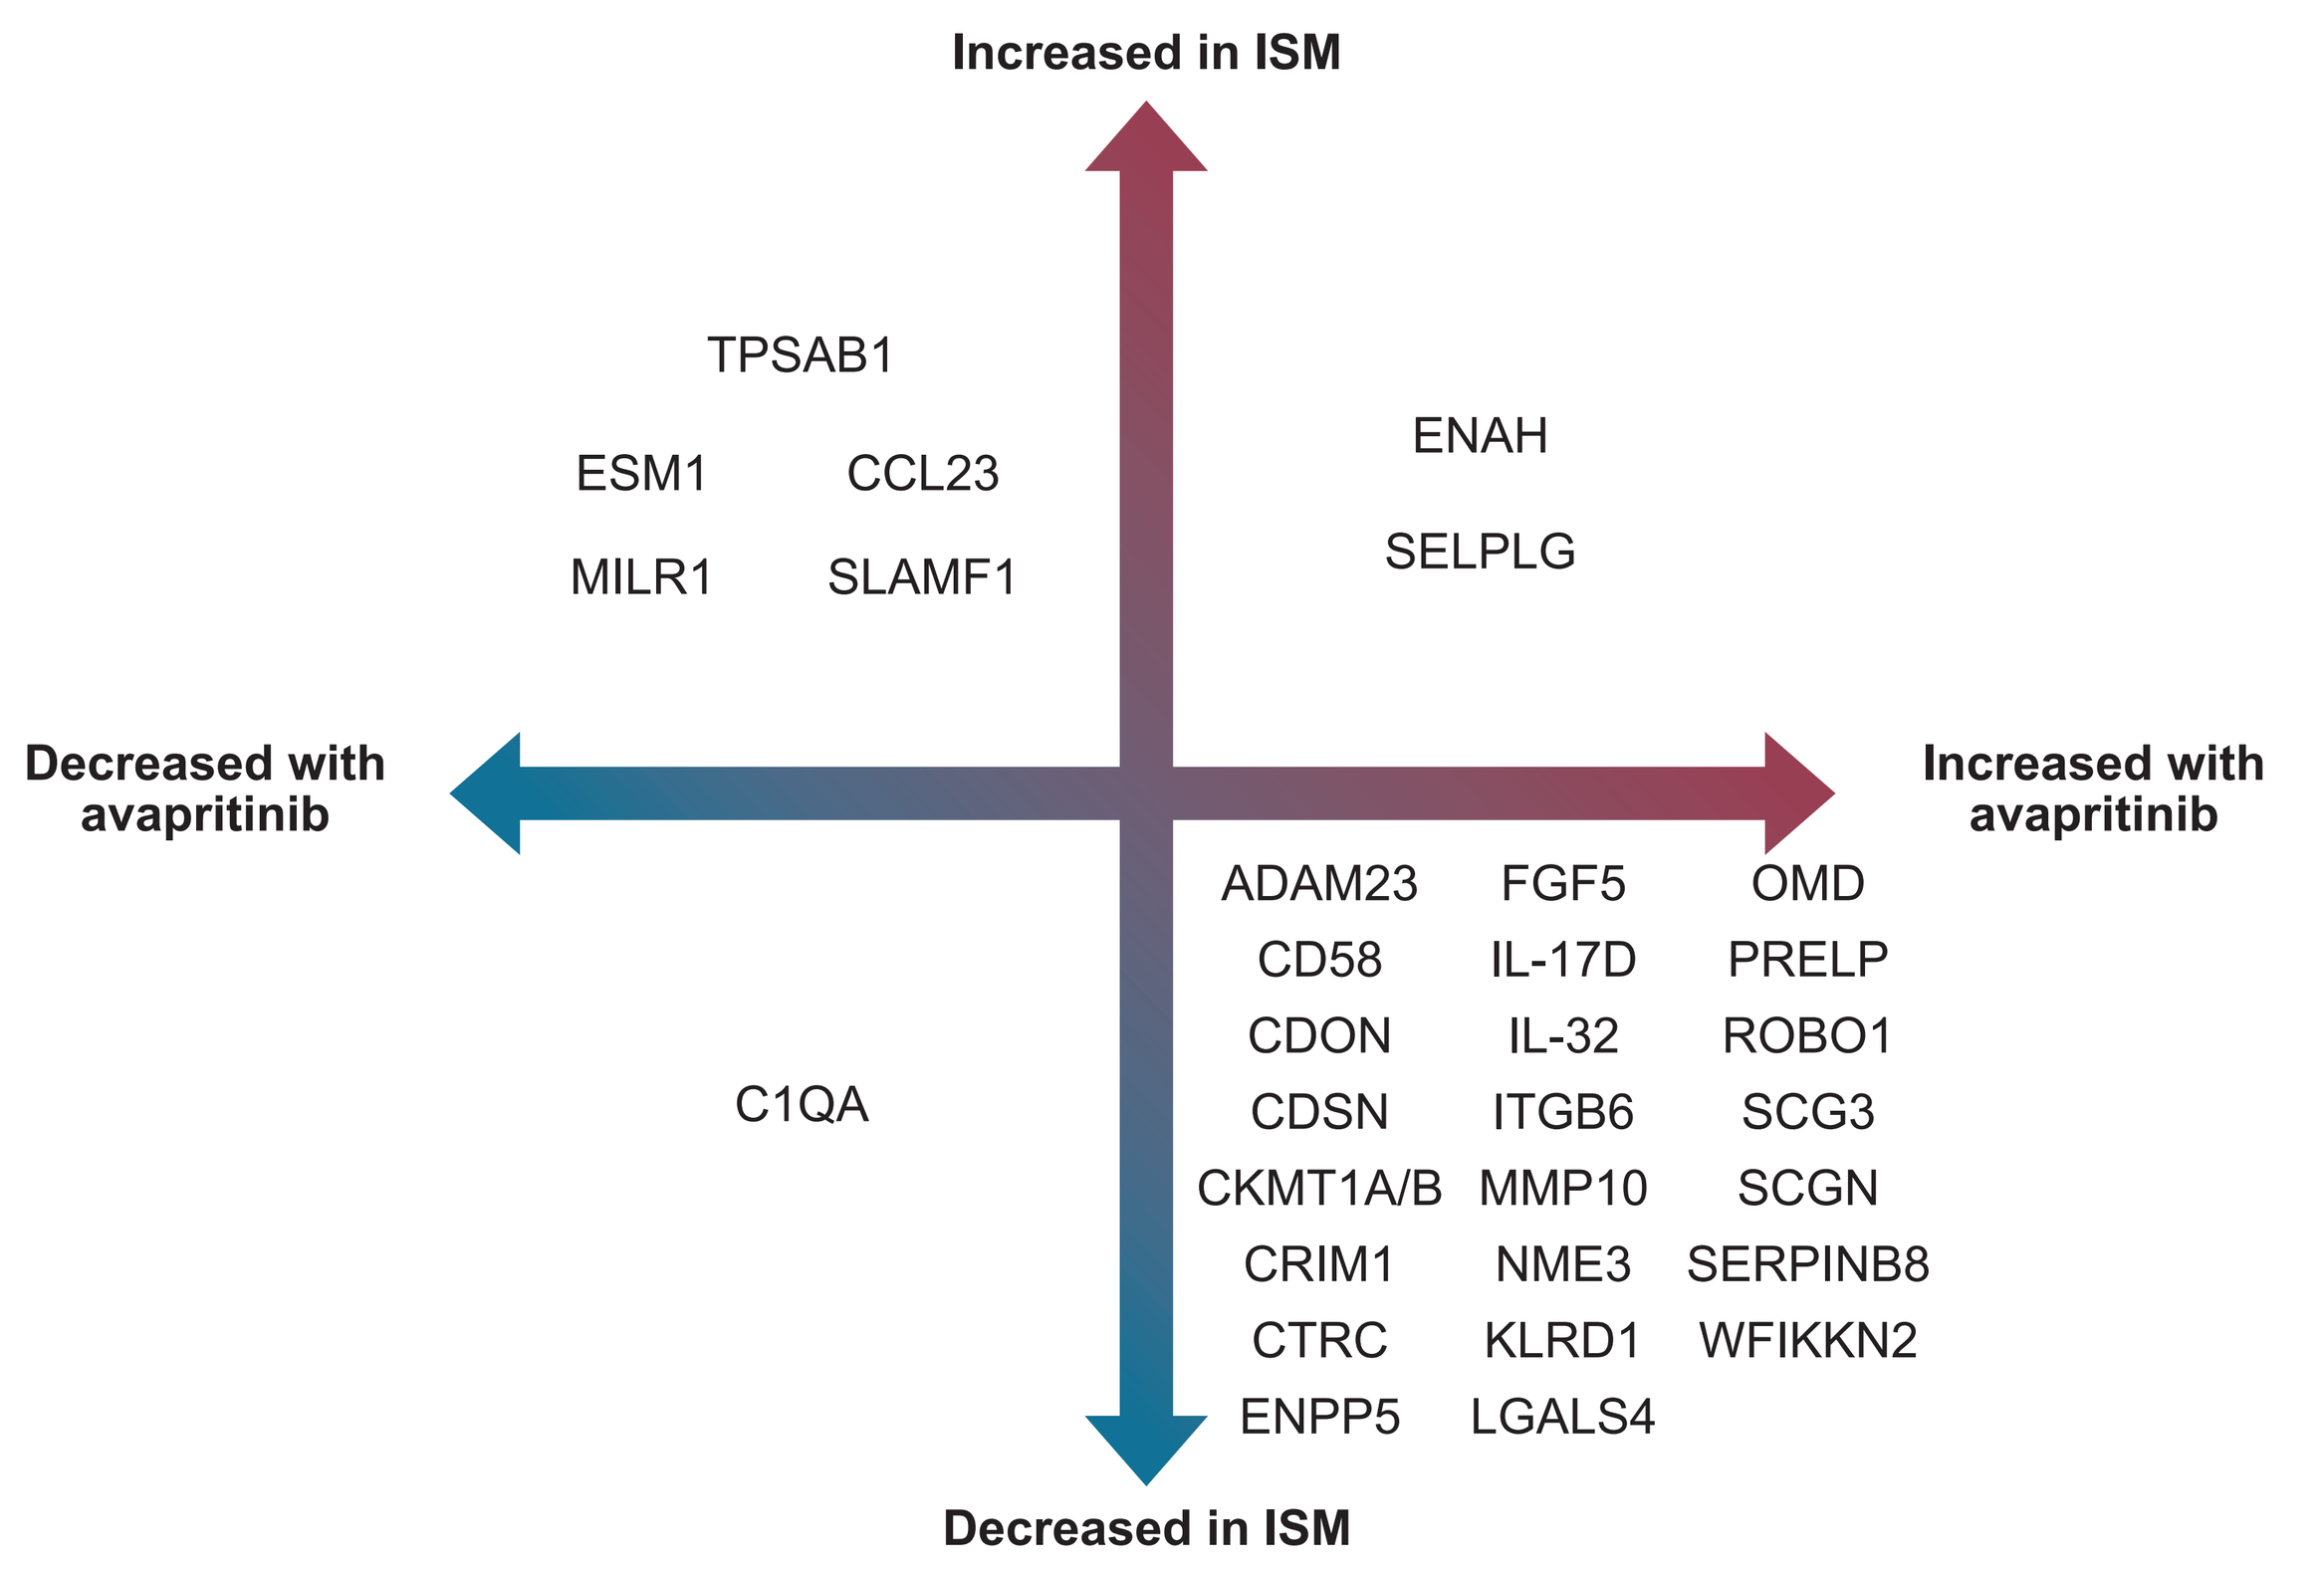

Increased in ISM
TPSAB1
ESM1
CCL23
MILR1
SLAMF1
ENAH
SELPLG
Decreased with avapritinib
Increased with avapritinib
C1QA
ADAM23
CD58
CDON
CDSN
CKMT1A/B
CRIM1
CTRC
ENPP5
FGF5
IL-17D
IL-32
ITGB6
MMP10
NME3
KLRD1
LGALS4
OMD
PRELP
ROBO1
SCG3
SCGN
SERPINB8
WFIKKN2
Decreased in ISM

Results

- Our analysis identified significantly higher levels of tryptase (TPSAB1) in patients with ISM *versus* healthy donors and also identified significant decreases in tryptase in patients receiving avapritinib, validating the ability of this technique to detect clinically meaningful differences in the levels of circulating plasma proteins
- Cytokine analysis identified multiple proteins previously linked to MC biology and SM. For example, CCL23 is produced by MCs and its levels correlate with the severity of disease, with the highest levels seen in patients with advanced SM[16,17]
 - Avapritinib significantly reduced the levels of elevated CCL23 seen in patients with ISM
- Out of the 31 proteins significantly changed by avapritinib, 28 were changed towards normal levels (**Figure 5**)

Conclusions

- Many differences are seen between the plasma proteome of patients with ISM *versus* healthy individuals, highlighting the immune dysregulation in this disease
- This analysis provides insight into the effects of avapritinib treatment on the inflammatory plasma proteome
- We provide evidence that circulating inflammatory proteins could serve as biomarkers of ISM disease burden and also may identify new potential therapeutic targets for its treatment

References

1. Bergström A et al. *Acta Oncol*. 2024;63:44–50; 2. Kristensen T et al. *Am J Hematol*. 2014;89:493–498; 3. Ungerstedt J et al. *Cancers*. 2014;14:3942; 4. Garcia-Montero AC et al. *Blood*. 2006;108:2366–2372; 5. Pardanani A. *Am J Hematol*. 2023;98:1097–1116; 6. Mesa RA et al. *Cancer*. 2022;128:3691–3699; 7. Hermine O et al. *PLoS One*. 2008;3:e2266; 8. van Anrooij B et al. *Allergy*. 2016;71:1585–1593; 9. Hartmann K et al. *J Allergy Clin Immunol*. 2016;137:35–45; 10. Akin C et al. *J Allergy Clin Immunol*. 2022;149:1912–1918; 11. Evans EK et al. Sci Transl Med. 2017;9:e1690; 12. Tashi T et al. Annual Meeting of the American Society of Hematology 2023, Oral #76; 13. Gotlib J et al. *NEJM Evidence*. 2023;2:EVIDoa2200339; 14. Ayvakit (avapritinib). Prescribing Information. Cambridge, MA: Blueprint Medicines Corporation; 2024; 15. Ayvakyt (avapritinib) Summary of Product Characteristics. Cambridge, MA; Blueprint Medicines Corporation; 2024; 16. Söderlund S et al. *J Allergy Clin Immunol*. 2023;152:205–213; 17. Mayado A et al. *Leukemia*. 2016;30:124–130.

Acknowledgements

Writing and editorial support under the direction of the authors was provided by Matt Nicolas, MSc, Danielle Scheunemann, MS, and Sarah Christopher, PhD, all of Paragon (a division of Prime, Knutsford, UK), funded by Blueprint Medicines Corporation

Poster available for download at:

Disclosures

Dr Akin has received consulting fees and research support from Blueprint Medicines Corporation, and Cogent Biosciences, and consulting fees from Novartis.

For all author disclosures, please contact medinfo@blueprintmedicines.com.

Presented at the 2025 AAAAI/WAO Joint Congress; February 28–March 3, 2025, San Diego, CA. Please contact medinfo@blueprintmedicines.com for permission to reprint and/or distribute.